Poster No: FP.27

Results of a double-blind cross-over trial of vamorolone in Duchenne muscular dystrophy (DMD): an alternative to traditional corticosteroids

Hoffman EP,[1] Clemens PR,[2] Perlman SJ,[3] Smith EC,[4] Horrocks I,[5] Finkel RS,[6] Mah JK,[7] Deconinck N,[8] Goemans N,[9] Haberlova J,[10] Straub V,[11] Ryan MM,[12] Tulinius M,[13] Webster R,[14] McMillan HJ,[15] Baranello G,[16] Spinty S,[17] Childs AM,[18] Selby KA,[19] Vilchez-Padilla JJ,[20] Nascimento-Osorio A,[21] Niks EH,[22] de Groot IJM,[23] Katsalouli M,[24] Guglieri M[11]

[1]ReveraGen BioPharma, Rockville, Maryland; [2]University of Pittsburgh School of Medicine, Pittsburgh, PA, USA; [3]University of Washington School of Medicine, Seattle, WA, USA; [4]Duke University School of Medicine, Durham, NC, USA; [5]Royal Hospital for Children, Glasgow, UK; [6]Nemours Children's Hospital, Orlando, FL, USA; [7]Alberta Children's Hospital Research Institute, Calgary, AB, Canada; [8]Neuromuscular Reference Center, Hôpital Universitaire des Enfants Reine Fabiola, Neurology, Brussels, Belgium; [9]KU Leuven, Leuven, Belgium; [10]Motol University Hospital, Prague, Czech Republic; [11]John Walton Muscular Dystrophy Research Centre, Newcastle University, Newcastle upon Tyne, UK; [12]The Royal Children's Hospital; [13]Queen Silvia Children's Hospital, Gothenburg, Sweden; [14]The Children's Hospital at Westmead, Sydney, NSW, Australia; [15]Children's Hospital of Eastern Ontario, Pediatric Neurology, Ottawa, ON, Canada; [16]The Dubowitz Neuromuscular Centre, UCL Great Ormond Street Institute of Child Health, London, UK; [17]Alder Hey Children's NHS Foundation Trust, Liverpool, UK; [18]Leeds Teaching Hospitals Trust, Leeds, UK; [19]BC Children's Hospital Research Institute, Vancouver, Canada; [20]Hospital Quirónsalud Valencia, Valencia, Spain; [21]Neuropaediatrics Department, Institut de Recerca Pediàtrica Hospital, Barcelona, Spain; [22]Leiden University Medical Center, Leiden, Netherlands; [23]UMC St Radboud, Nijmegen, Netherlands; [24]"P&A Kyriakou" Children's Hospital, Athens, Greece.

Introduction

- Vamorolone is an investigational dissociative steroidal anti-inflammatory drug that seeks to retain efficacy and potentially reduce select safety concerns in patients with Duchenne muscular dystrophy (DMD) compared to corticosteroids via changes to structure/activity relationships with the glucocorticoid receptor.[1]
- A series of open-label studies (VBP15-LTE, NCT03038399) in boys with DMD suggested a favorable efficacy–safety profile over 30 months of exposure.[2]
- The efficacy and safety of vamorolone were investigated during the first 24-weeks (Period 1) of the VISION-DMD (VBP15-004, NCT03439670) study:
 - The results of the primary analysis at 24 weeks have been reported previously.[3]
 - The study met its primary endpoint; both doses of vamorolone (6 mg/kg/day and 2 mg/kg/day) showed statistically significant and clinically meaningful improvement in functional outcomes vs. placebo after 24 weeks of treatment.
 - Height percentile declined in prednisone-treated, but not vamorolone-treated participants.
 - Bone turnover markers declined with prednisone treatment but not vamorolone.

Objective

- Evaluate the efficacy and safety of continuous 48-week vamorolone treatment.
- Assess efficacy of VAM 6mg/kg in patients switching from PDN 0.75 mg/kg in period 2 of the study.

Methods

- VISION-DMD (VBP15-004) is a 48-week randomized, double-blind study comprising two periods.
 - During Period 1, 121 patients were randomized 1:1:1:1 to vamorolone 2 or 6 mg/kg/day, prednisone 0.75 mg/kg/day, or placebo for 24 weeks.
 - During Period 2, patients continued their initial vamorolone dose or crossed over from placebo or prednisone to vamorolone 2 or 6 mg/kg/day.
- The patients were 4–<7 years of age at baseline with centrally confirmed DMD who could walk independently without assistive devices and complete the time to stand from supine (TTSTAND) test without assistance in <10 seconds at baseline. The patients were steroid-naïve at baseline.
- This analysis is conducted in patients who were randomized to receive vamorolone 2 or 6 mg/kg/day throughout the 48-week study, modified intention to treat-2 population (**Figure 1**) as well as evaluate the impact of efficacy on patients switched to VAM 6mg/kg in treatment period 2.
- Participants who did not have an efficacy measurement in period 2 were excluded (All randomized subjects who had at least one dose of study medication and had at least one post-baseline efficacy assessment during Period 2 were included; this was the primary analysis population at Week 48)
- Global efficacy was assessed as change from baseline to week 48 in time to stand (TTSTAND) velocity, 6-minute walk distance (6MWT), Time to run/walk 10 m (TTRW) velocity, North Star Ambulatory Assessment (NSAA) score, Time to climb 4 steps (TTCLIMB) velocity and was modelled using restricted maximum likelihood-based mixed model for repeated measures (MMRM).

Figure 1: Study design for VISION-DMD (VBP15-004).

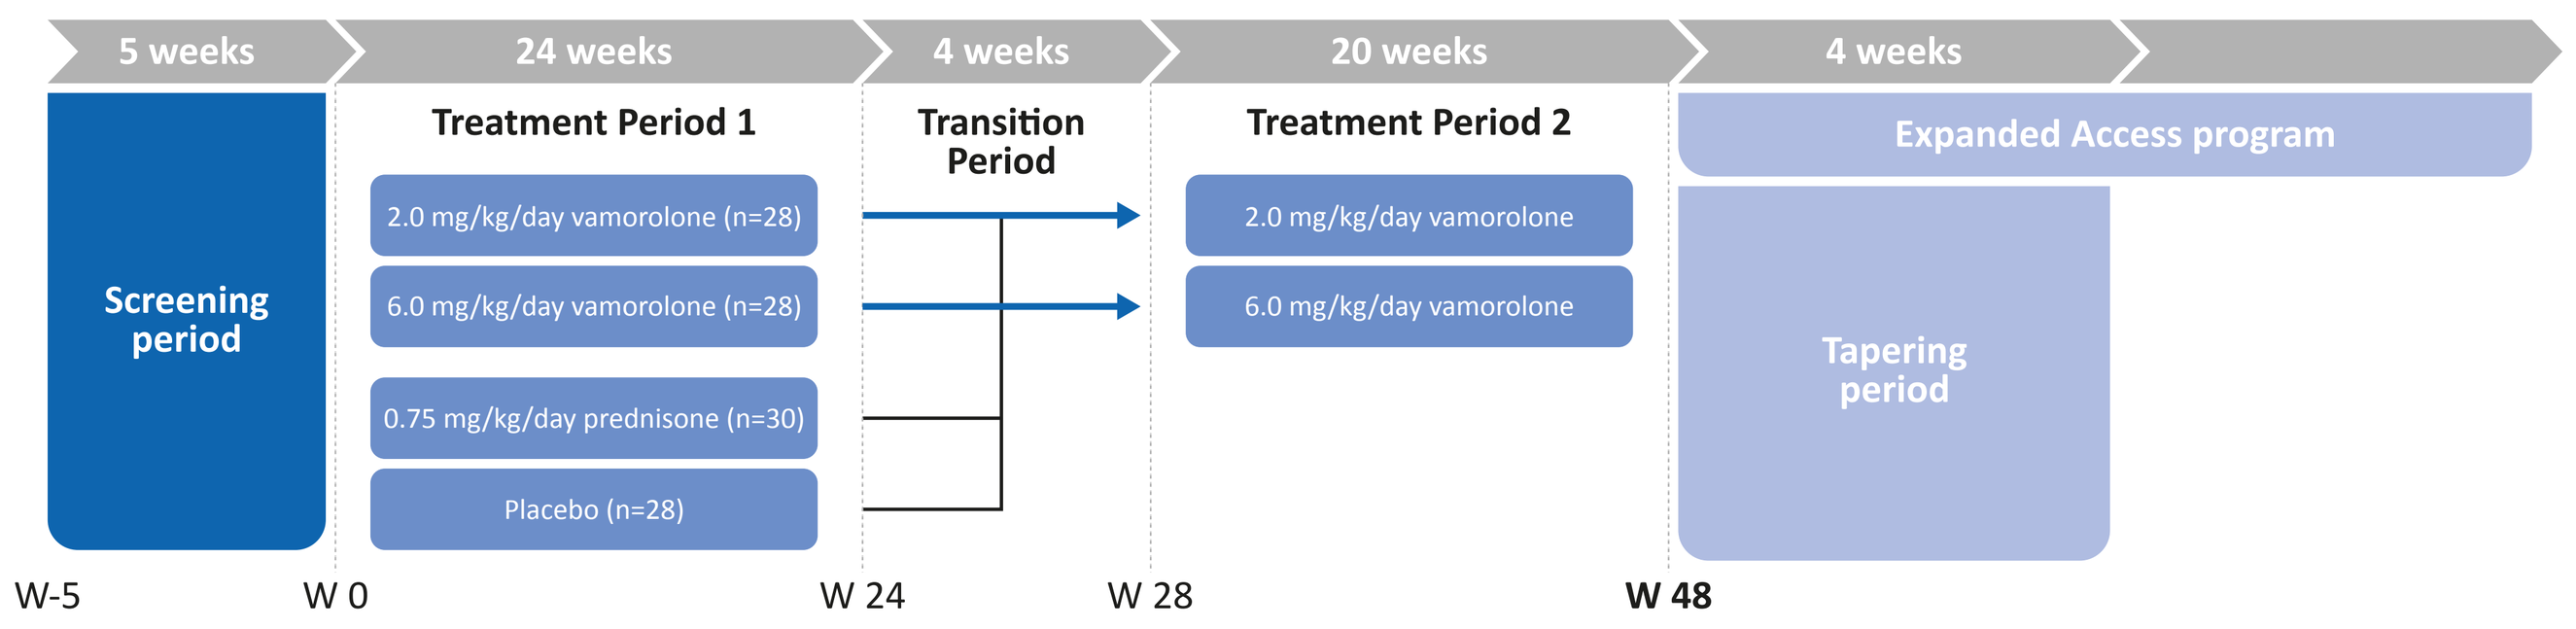
5 weeks
24 weeks
4 weeks
20 weeks
4 weeks
Screening period
Treatment Period 1
2.0 mg/kg/day vamorolone (n=28)
6.0 mg/kg/day vamorolone (n=28)
0.75 mg/kg/day prednisone (n=30)
Placebo (n=28)
Transition Period
Treatment Period 2
2.0 mg/kg/day vamorolone
6.0 mg/kg/day vamorolone
Expanded Access program
Tapering period
W-5
W 0
W 24
W 28
W 48

Results

- Of the 121 patients randomized to the study, 56 received vamorolone during Periods 1 and 2.
 - Two of 56 patients discontinued treatment during Period 2 (1 adverse event [AE], 1 consent withdrawn).
- Thirty patients received prednisone during Period 1 followed by vamorolone treatment during Period 2.
 - All 30 patients completed vamorolone treatment at Week 48.
- Baseline characteristics were similar for age, although differences were noted in favor of PDN for select efficacy variables as well as height, weight, and BMI.
- For the primary endpoint, TTSTAND velocity, the effect seen at Week 24 for the continuous vamorolone 6 mg/kg group was maintained until Week 48, with a significant difference vs. continuous vamorolone 2 mg/kg at Week 48 (p=0.001) (**Figure 2**).
- For the continuous vamorolone groups, in the other secondary efficacy endpoints, vamorolone 6 mg/kg/day showed greater improvements in motor outcomes compared to 2 mg/kg/day for 6MWT (p=0.047) and TTCLIMB (p=0.031), while similar efficacy was observed for TTRW (p=0.375) and NSAA (p=0.602) (**Figure 3**).
- Prednisone-treated participants (Period 1) that crossed over to vamorolone showed maintenance of efficacy across all efficacy endpoints for vamorolone 6 mg/kg/day: TTSTAND velocity (0.05 vs 0.04 rises/sec, compared to baseline), 6-MWT (40 vs 29 m), TTRW velocity (0.26 vs 0.19 m/sec), NSAA (4.7vs 4.0 points) and TTCLIMB (0.07 vs 0.08 tasks/sec).
 - These changes were similar in magnitude to that seen with continuous treatment with vamorolone 6 mg/kg/day throughout the study.
 - For the vamorolone 2 mg/kg/day group, statistically significant differences were seen vs. placebo at Week 24, but overall, the effect was lower than for vamorolone 6 mg/kg/day or prednisone 0.75 mg/kg/day and not consistently maintained for all efficacy endpoints.
- Placebo-treated participants in Period 1 that crossed over to vamorolone in Period 2 (delayed starters) showed an improvement in multiple efficacy outcomes after the switch to vamorolone.
 - Improvement in multiple efficacy outcomes in delayed starters post-crossover to vamorolone 6.0 mg/kg/day were observed (TTSTAND velocity, TTCLIMB velocity, 6MWD, NSAA; all p<0.05).
- The most common adverse event of special interest (AESIs) in Period 1 were Infections, GI symptoms, Cushingoid features and Behavior problems:
 - Infections (vamorolone 2 mg/kg/day: 43.3% [n=13]; vamorolone 6 mg/kg/day: 32.1% [n=9]; placebo: 44.8% [n=13]; prednisone: 38.7% [n=12])
 - GI symptoms (vamorolone 2 mg/kg/day: 30.3% [n=9]; vamorolone 6 mg/kg/day: 28.6% [n=8]; placebo: 27.6% [n=8]; prednisone: 25.8% [n=7])
 - Cushingoid (vamorolone 2 mg/kg/day: 6.7% [n=2]; vamorolone 6 mg/kg/day: 28.6% [n=8]; placebo: 0%; prednisone: 22.6% [n=7])
 - Behavior problems (vamorolone 2 mg/kg/day: 16.7% [n=5]; vamorolone 6 mg/kg/day: 21.4% [n=6]; placebo: 13.8% [n=4]; prednisone: 31.8% [n=10])
- During the 48 weeks, three serious AEs were reported in the continuous vamorolone groups: perforated appendicitis (6 mg/kg/day), asthma (6 mg/kg/day), and viral gastroenteritis (2 mg/kg/day), all considered unrelated to vamorolone.
- No serious adverse events (AEs) were reported after the switching from prednisone to either vamorolone dose.
- The frequency of AEs (all AEs adjusted by duration of treatment) was reduced after the switch from prednisone to vamorolone (**Figure 4**).
 - Out of all AESIs, the largest reductions in total AEs by patient/year were seen in behavior problems (prednisone to vamorolone: 1.1 to 0.5) and gastrointestinal symptoms (prednisone to vamorolone: 0.75 to 0.6).
 - No subject switching from prednisone reported Cushingoid features on vamorolone 6 mg/kg/day.
- Body mass index (BMI) stabilized for the continuous vamorolone 6 mg/kg group in Period 2 after an initial increase observed during the first 24 weeks BMI remained stable in subjects switching from prednisone to vamorolone 6 mg/kg/day.
- No stunting of growth was observed with either vamorolone dose in the continuous vamorolone groups.
 - The average baseline height indicated a height lower than in the general age-matched population in keeping with known natural history (**Table 1**).
 - Mean (SD) change from baseline to week 48 for vamorolone 2 and 6 mg/kg/day was 0.13 (0.277) and 0.29 (0.355) in contrast to stunting of growth known to occur with corticosteroids.[4]
- Stunting of growth observed with prednisone during Period 1 was reversed during treatment with vamorolone during Period 2 (**Figure 5**).
- Decreases in serum bone biomarkers (osteocalcin, procollagen I N-terminal propeptide [PINP]) and collagen I C-Telopeptide [CTX], and alkaline phosphatase [ALP]) seen in the prednisone group during Period 1 were quickly reversed after the switch to vamorolone during Period 2.

Table. Characteristics at baseline (study entry), mITT-2 / Safety-2 population

	Continuous vamorolone 2 mg/kg/day (n=28) Mean (SD)	Continuous vamorolone 6mk/kg/day (n=28) Mean (SD)	Prednisone to vamorolone 2 mg/kg/day (n=15) Mean (SD)	Prednisone to vamorolone 6 mg/kg/day (n=15) Mean (SD)
Age (years)	5.3 (0.9)	5.4 (0.9)	5.4 (0.9)	5.6 (0.8)
TTSTAND (sec)	6.0 (2.4)	6.0 (2.0)	5.4 (1.9)	4.5 (0.8)
6-MWT (m)	317 (60)	313 (56)	329 (52)	360 (58)
NSAA (points)	17.5 (4.6)	18.9 (4.1)	21.1 (5.4)	21.4 (5.8)
Height (percentile)	32.0 (29.2)	23.2 (24.6)	34.2 (30.1)	41.1 (29.1)
Weight (percentile)	43.1 (29.0)	43.7 (26.7)	53.0 (28.7)	61.1 (28.2)
BMI (percentile)	63.5 (27.9)	69.8 (23.0)	73.6 (24.0)	77.4 (17.5)

6MWD, six-minute walk distance; BMI, body mass index; mITT-2, modified intention to treat-2 population; MMRM, mixed model for repeated measures; TTRW, time to run/walk 10 m; NSAA, North Star Ambulatory Assessment; SD, standard deviation.

Figure 2. TTSTAND velocity (rises/sec) (mITT-2 population, MMRM)

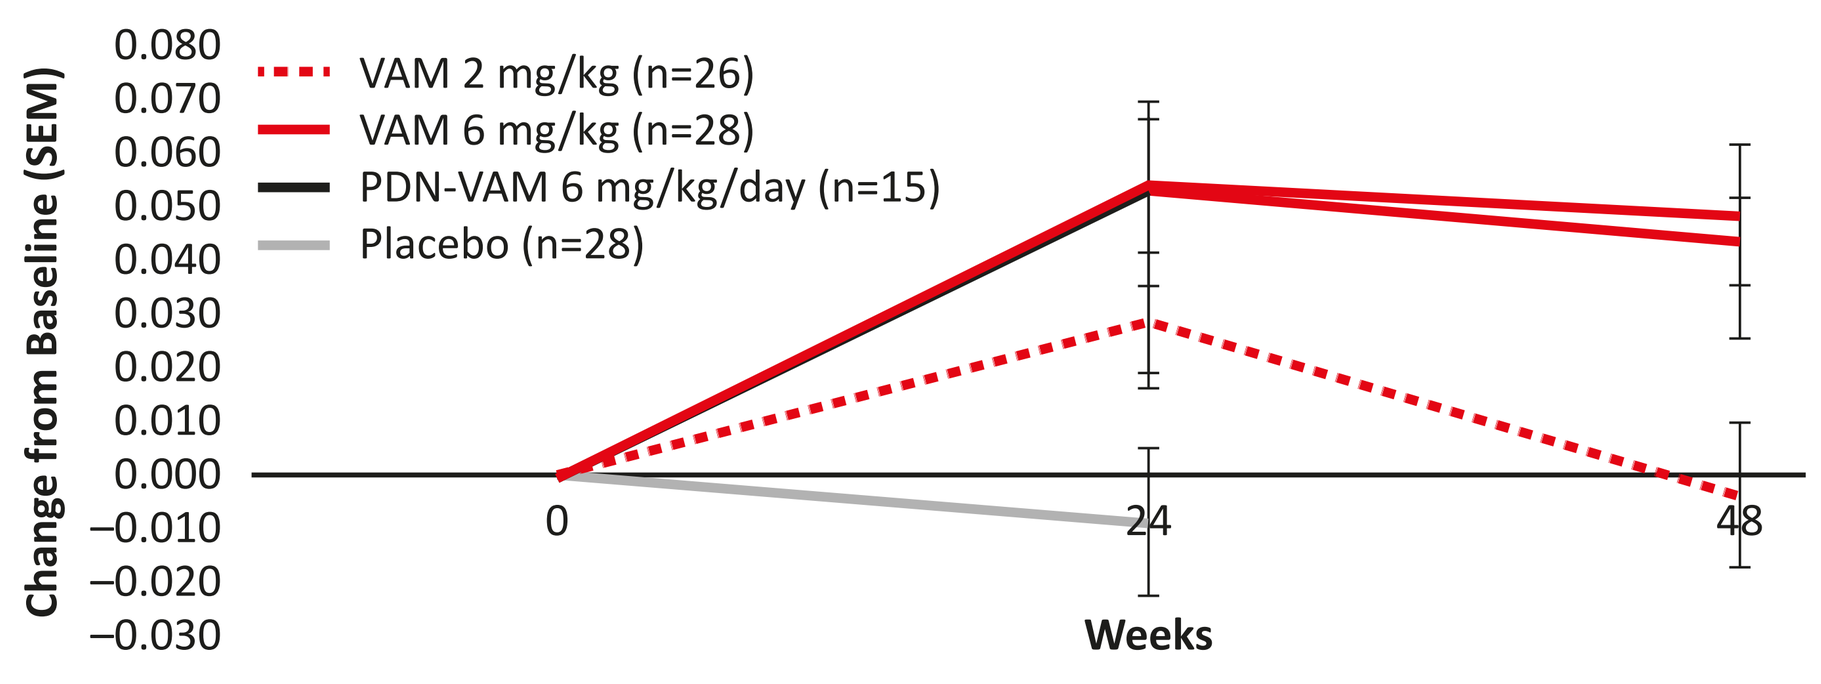
VAM 2 mg/kg (n=26)
VAM 6 mg/kg (n=28)
PDN-VAM 6 mg/kg/day (n=15)
Placebo (n=28)
Change from Baseline (SEM)
0.080
0.070
0.060
0.050
0.040
0.030
0.020
0.010
0.000
−0.010
−0.020
−0.030
0
24
48
Weeks

mITT-2, modified intention to treat-2 population; MMRM, mixed model for repeated measures; SEM, standard error of the mean; TTSTAND, time to stand from supine.

Figure 3. 6MWT distance (m), TTRW velocity (m/sec), NSAA score (points) and TTCLIMB velocity (tasks/s) (mITT-2 population, MMRM)

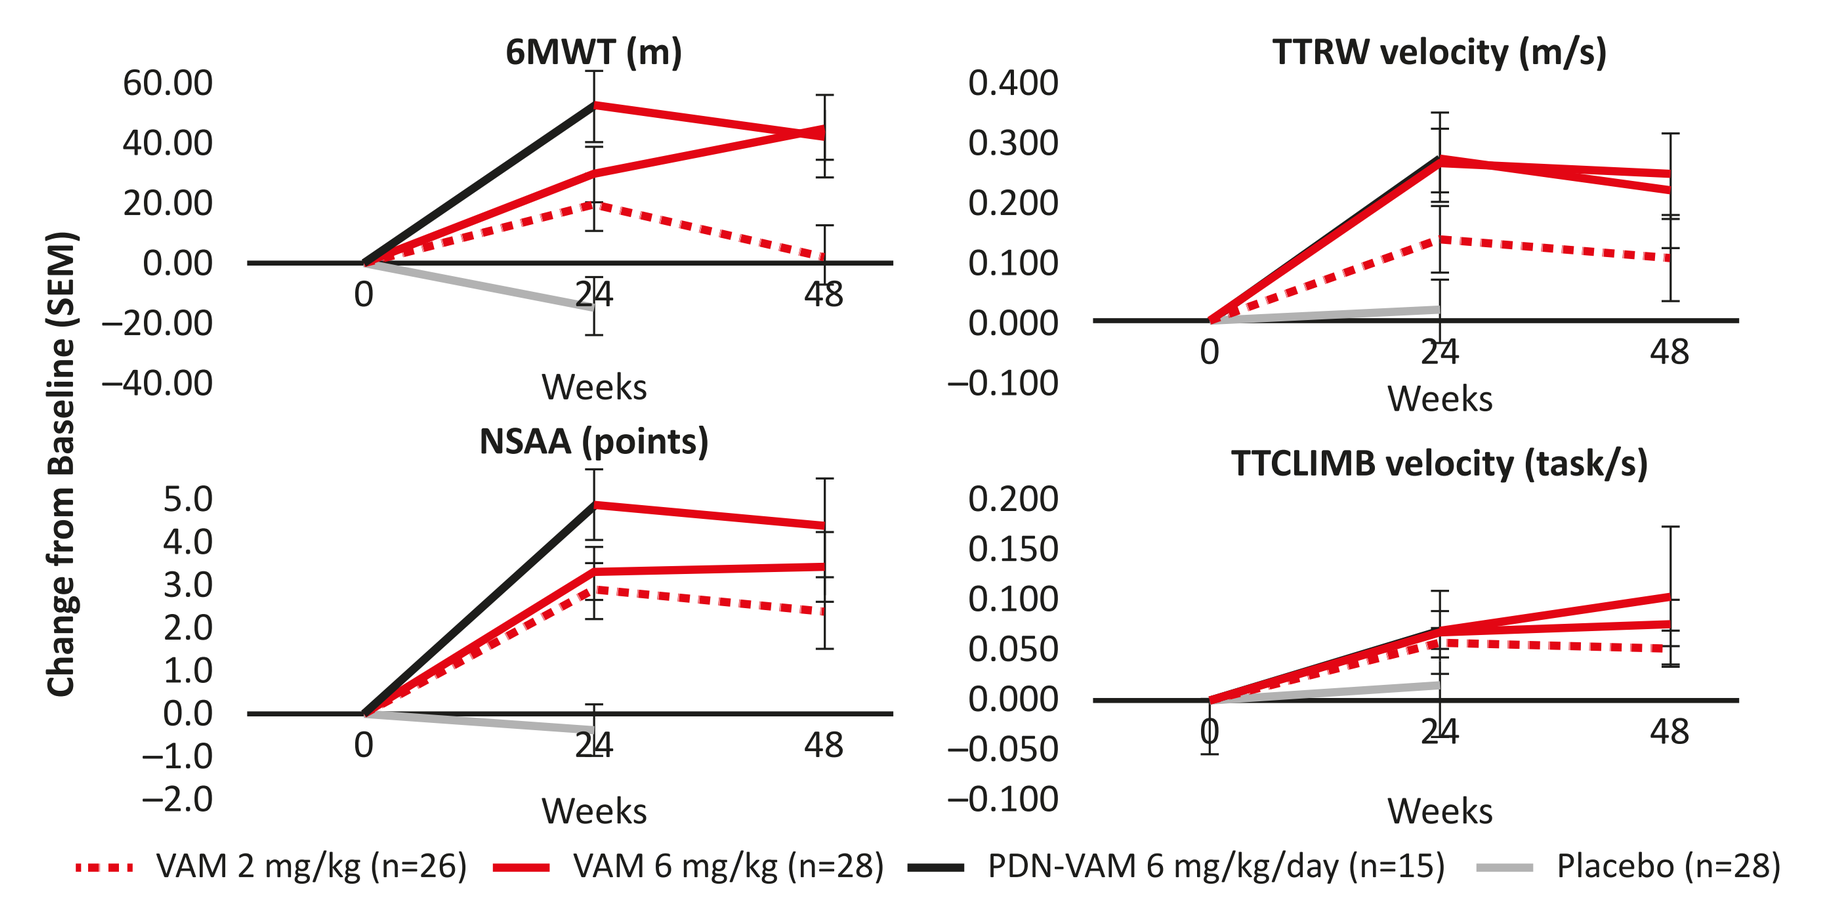
6MWT (m)
TTRW velocity (m/s)
NSAA (points)
TTCLIMB velocity (task/s)
Change from Baseline (SEM)
Weeks
0
24
48
VAM 2 mg/kg (n=26)
VAM 6 mg/kg (n=28)
PDN-VAM 6 mg/kg/day (n=15)
Placebo (n=28)

6MWD, six-minute walk distance; mITT-2, modified intention to treat-2 population; MMRM, mixed model for repeated measures; TTCLIMB, time to climb 4 steps; TTRW, time to run/walk 10 m; NSAA, North Star Ambulatory Assessment; SEM, standard error of the mean.

Figure 4. Rates of all TEAEs and AESIs, in patients who switched from prednisone 0.75 mg/kg/day to vamorolone 2 or 6 mg/kg/day (Safety-2 population)

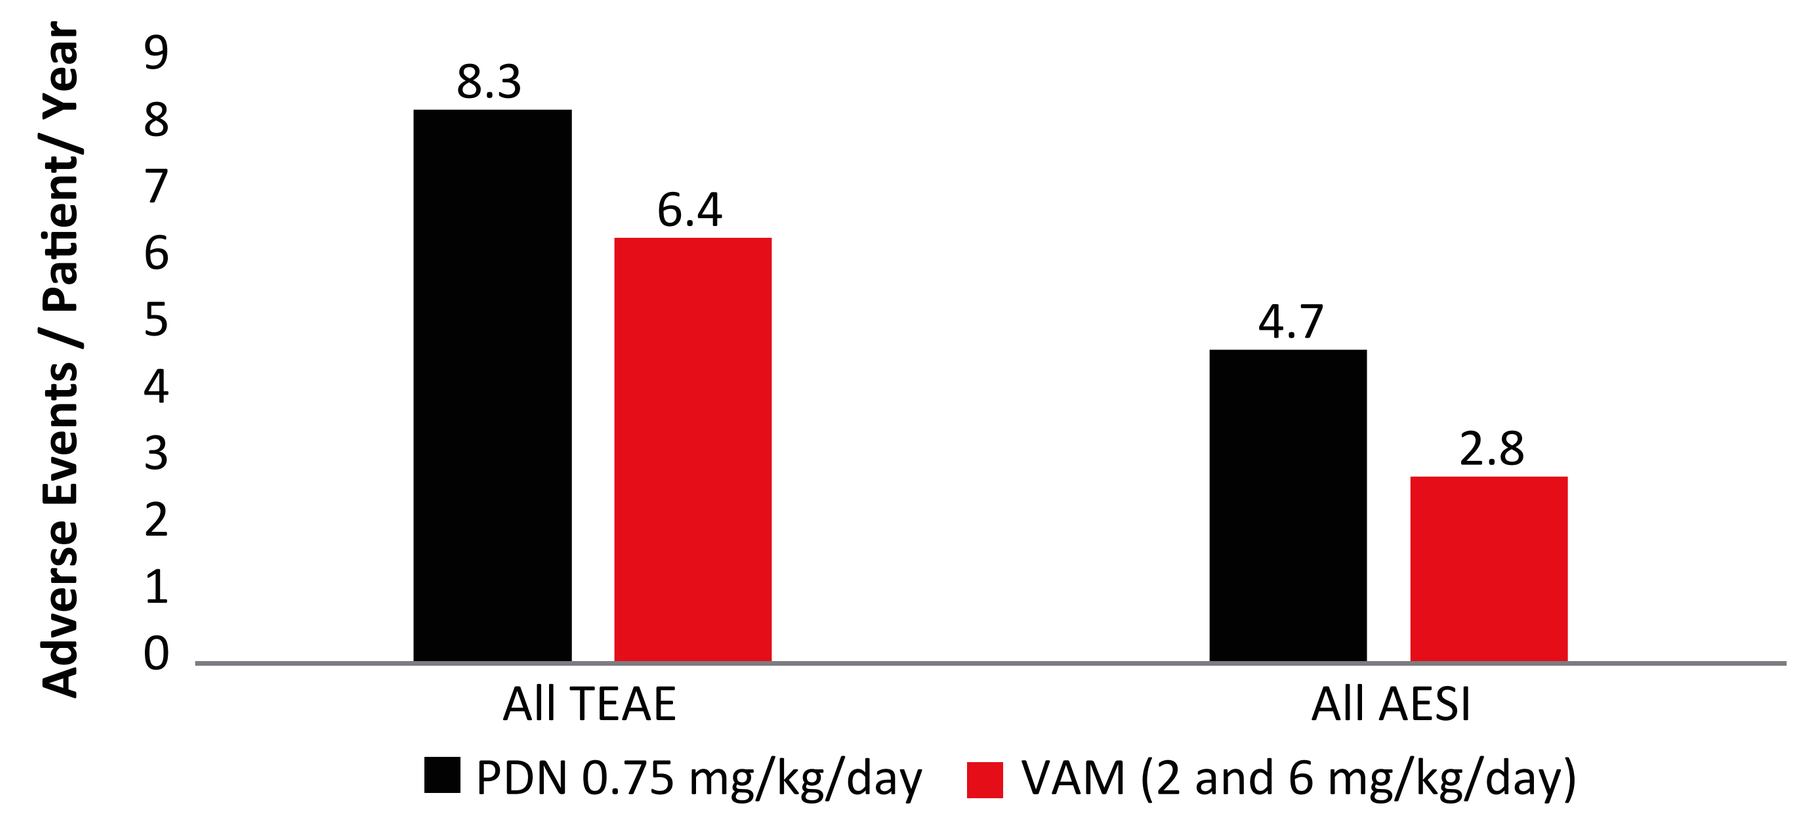
Adverse Events / Patient/ Year
8.3
6.4
4.7
2.8
All TEAE
All AESI
PDN 0.75 mg/kg/day
VAM (2 and 6 mg/kg/day)

AESI, adverse event of special interest; TEAE, treatment-emergent adverse event.

Figure 5: Change in height z-scores, in patients who switched from prednisone 0.75 mg/kg/day to vamorolone 2 or 6 mg/kg/day (Safety-2 population)

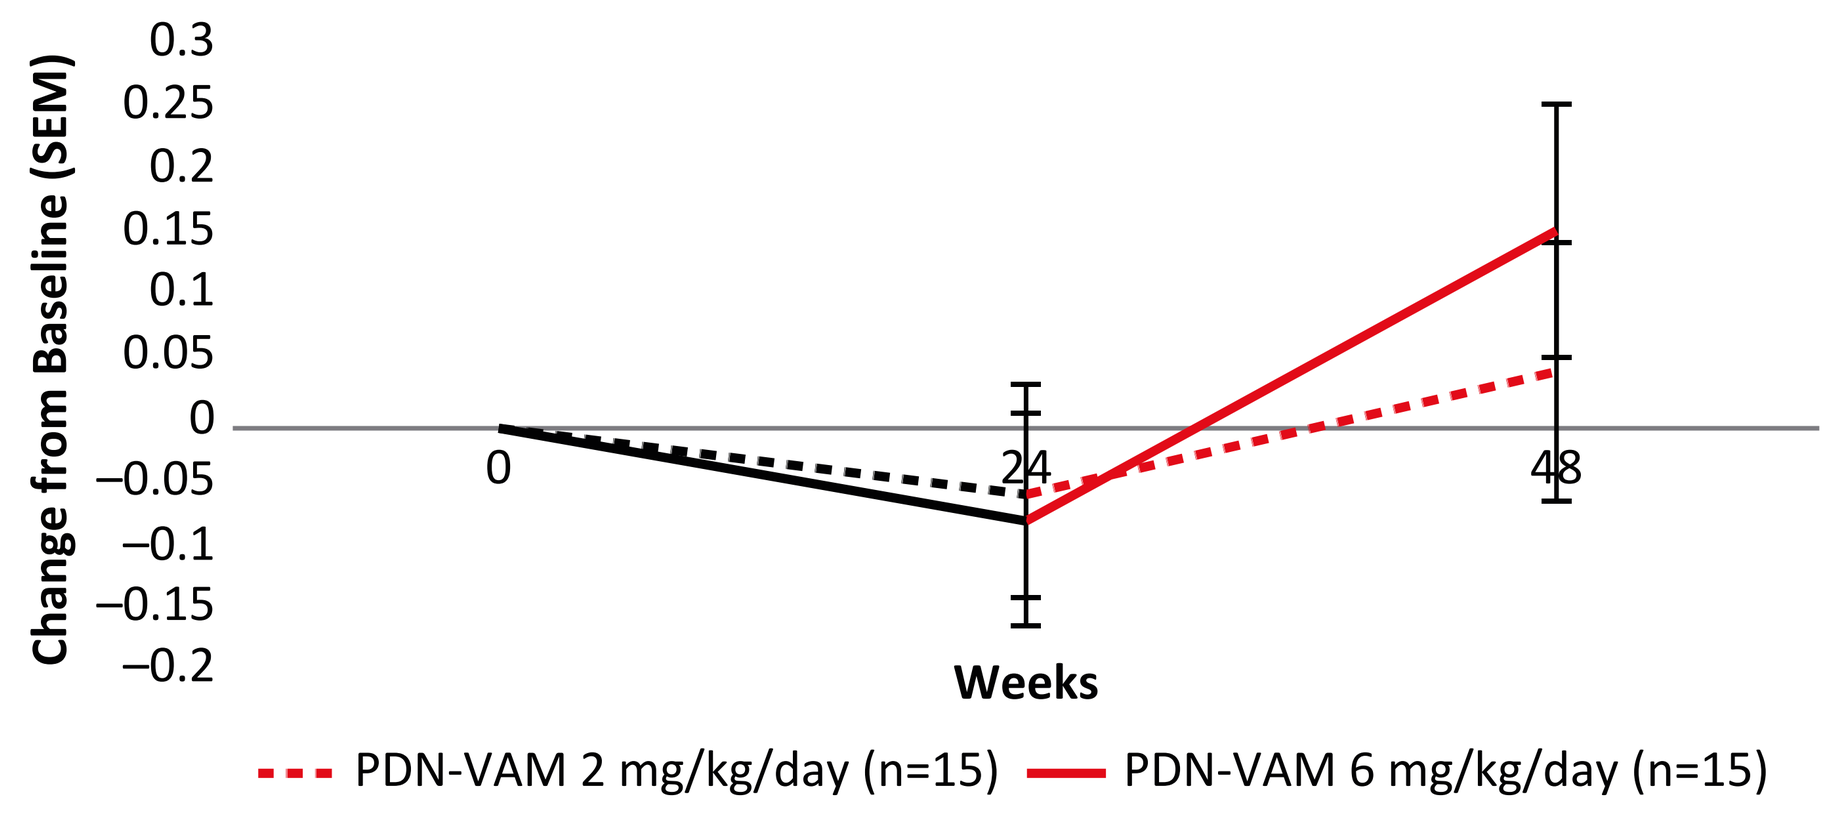
Change from Baseline (SEM)
0.3
0.25
0.2
0.15
0.1
0.05
0
−0.05
−0.1
−0.15
−0.2
0
24
48
Weeks
PDN-VAM 2 mg/kg/day (n=15)
PDN-VAM 6 mg/kg/day (n=15)

PDN, prednisone; SEM, standard error of the mean; VAM, vamorolone.

Conclusion

- For the continuous vamorolone groups, efficacy of vamorolone 6 mg/kg/day established at 24 weeks was maintained over 48 weeks across all outcome measures, while only across some measures for 2 mg/kg/day. There was no loss of efficacy after switching from prednisone to vamorolone 6 mg/kg/day.
- Vamorolone treatment was generally well tolerated at both continuous dose levels throughout 48 weeks and for patients who switched to vamorolone during Period 2.
- For subjects who continued on the same dose of vamorolone throughout the study, the safety profile was consistent at week 48 compared to the results previously reported at week 24.
- No stunting of growth was seen with either vamorolone dose, consistent with data previously presented from long-term open-label studies.
- Subjects switching from prednisone to vamorolone resumed normal growth, and experienced reductions in select other side effects, as well as reduced impact on bone biomarkers.
- BMI z-score stabilized in Period 2 in the continuous vamorolone 6 mg/kg group.
- The longer-term results of the VISION-DMD (VBP15-004) study confirm earlier findings regarding the sustained efficacy with a differentiated safety profile in patients switched from prednisone to vamorolone and presents data on switching to vamorolone following treatment with prednisone.

Acknowledgments

We would like to thank all patients with DMD and their families for participating in the research studies and the study coordinators, clinical evaluators, and Therapeutic Research in Neuromuscular Disorders Solutions.

References

1. Heier CR, et al. *Life Sci Alliance.* 2019;2(1):e201800186
2. Mah JK, et al. *JAMA Netw Open.* 2022 Jan 4;5(1):e2144178.
3. Hoffman E. Presented at PPMD Annual Conference, June 22–26, 2021 (Virtual)
4. Bello L, et al. *Neurology*. 2015;85: 1048–1055.