EP.147

2.5-years of vamorolone treatment in Duchenne Muscular Dystrophy: Results of an open label long-term extension

Hoffman, E.[1]; Dang, U.[2]; Clemens, P.[3]; Gordish-Dressman, H.[4]; Schwartz, B.[5]; Mengle-Gaw, L.[5]; Leinonen, M.[6]; Smith, E.[7]; Castro, D.[8]; Kuntz, N.[9]; Finkel, R.[10]; Tulinius, M.[11]; Nevo, Y.[12]; Ryan, M.[13]; Webster, R.[14]; van den Anker, J.[4]; Ward, L.[15]; Damsker, J.[16]; McDonald, C.[17]; Guglieri, M.[18]; Mah, J.[19]

1. Binghamton University - State University of New York, Pharmaceutical Sciences, Binghamton, USA; 2. Binghamton University – Health Outcomes, Binghamton, USA; 3. University of Pittsburgh, Pittsburgh, USA ; 4. Children's National Medical Center, District of Columbia, USA; 5. Camden Associates, St. Louis, USA; 6. Santhera, Pratteln, Switzerland; 7. Duke University, Chapel Hill, USA; 8. UTSW, Dallas, USA; 9. Lurie's Children's Hospital, Chicago, USA; 10. Nemours Children's Hospital, , Orlando, USA; 11. Queen Silvia Children's Hospital, Gothenburg, Sweden; 12. Tel Aviv University, Tel Aviv, Israel; 13. Royal Children's Hospital, Melbourne, Australia; 14. The Children's Hospital at Westmead, Sydney, Australia; 15. Children's Hospital of Eastern Ontario, Ottawa, Canada; 16. ReveraGen, Rockville, USA; 17. University of California Davis, Davis, USA; 18. University of Newcastle, Newcastle, UK; 19. Alberta Children's Hospital, Calgary, Canada

Background

- Vamorolone is a first-in-class dissociative steroid engineered to uncouple anti-inflammatory effects from glucocorticoid (GC)-mediated adverse effects[1]
- Growth delay is a well-recognized adverse event (AE) associated with traditional GCs, which are the standard-of-care (SOC) for patients with Duchenne Muscular Dystrophy (DMD)[1]
- Results from the Phase IIa 4-week (VBP15-002, NCT02760264) multiple ascending dose study of vamorolone in GC-naïve DMD boys aged 4 to <7 years have previously been published[2]
- A Phase IIa 24-week (VBP15-003, NCT02760277) open-label extension dose-finding study suggested efficacy of vamorolone treatment at 2.0 and 6.0 mg/kg/day based on motor outcomes[3]
- Interim analysis at 18 months of the vamorolone open-label long-term extension (LTE) study (VBP15-LTE, NCT03038399) showed ongoing improvement in motor function and a favorable safety profile[4]
- Here we report on the long-term safety, tolerability, and efficacy of vamorolone in boys with DMD who completed the 24-week VBP15-003 trial and the 24-month VBP15-LTE trial, with a total of up to 30 months of vamorolone treatment

Trial participants

- Three consecutive multicenter open-label clinical trials (VBP15-002/003/LTE) were conducted by the Cooperative International Neuromuscular Research Group (CINRG)
- A CONSORT Chart is shown **(Figure 1)**

Figure 1. CONSORT Chart

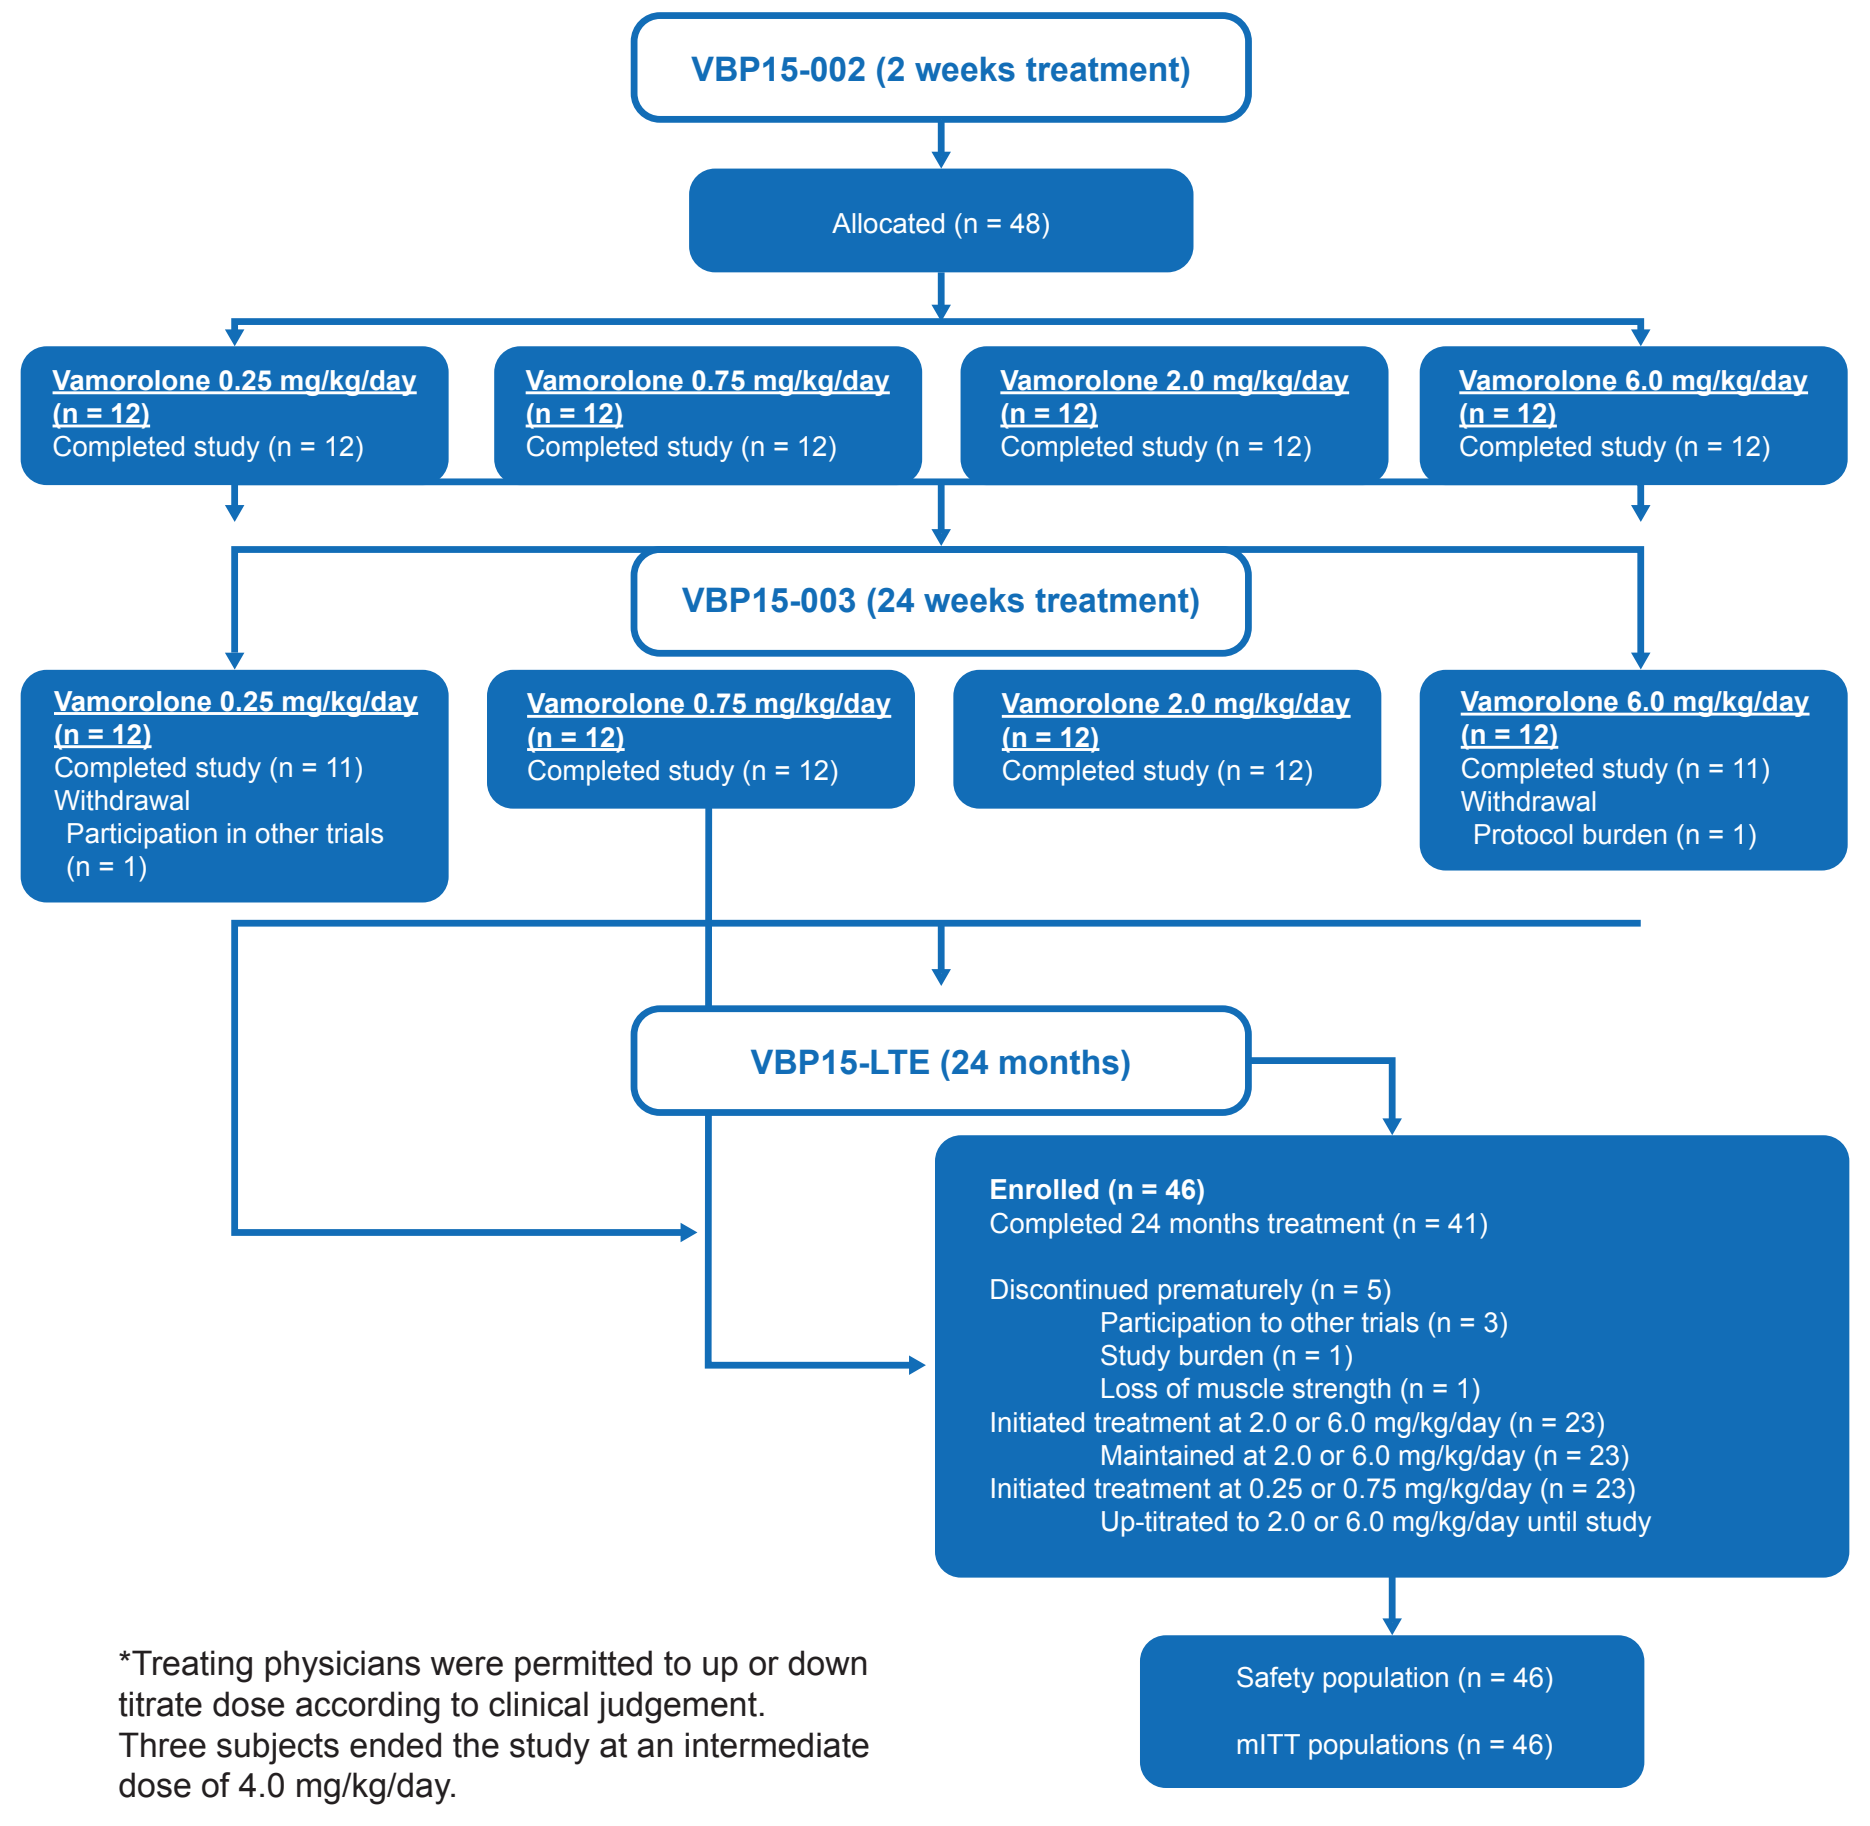

VBP15-002 (2 weeks treatment)
Allocated (n = 48)
Vamorolone 0.25 mg/kg/day (n = 12) Completed study (n = 12)
Vamorolone 0.75 mg/kg/day (n = 12) Completed study (n = 12)
Vamorolone 2.0 mg/kg/day (n = 12) Completed study (n = 12)
Vamorolone 6.0 mg/kg/day (n = 12) Completed study (n = 12)
VBP15-003 (24 weeks treatment)
Vamorolone 0.25 mg/kg/day (n = 12) Completed study (n = 11) Withdrawal Participation in other trials (n = 1)
Vamorolone 0.75 mg/kg/day (n = 12) Completed study (n = 12)
Vamorolone 2.0 mg/kg/day (n = 12) Completed study (n = 12)
Vamorolone 6.0 mg/kg/day (n = 12) Completed study (n = 11) Withdrawal Protocol burden (n = 1)
VBP15-LTE (24 months)
Enrolled (n = 46)
Completed 24 months treatment (n = 41)
Discontinued prematurely (n = 5)
Participation to other trials (n = 3)
Study burden (n = 1)
Loss of muscle strength (n = 1)
Initiated treatment at 2.0 or 6.0 mg/kg/day (n = 23)
Maintained at 2.0 or 6.0 mg/kg/day (n = 23)
Initiated treatment at 0.25 or 0.75 mg/kg/day (n = 23)
Up-titrated to 2.0 or 6.0 mg/kg/day until study
Safety population (n = 46)
mITT populations (n = 46)

*Treating physicians were permitted to up or down titrate dose according to clinical judgement. Three subjects ended the study at an intermediate dose of 4.0 mg/kg/day.

Results

Patients

- Most patients (89.1%) who entered the LTE completed the 2-year treatment period **(Figure 1)**
- Demographic and clinical characteristics of the vamorolone-treated participants were previously reported[4]
- Patient-level dose escalations are shown **(Figure 2)**

Efficacy

- Participants assigned to vamorolone at 2.0 and 6.0 mg/kg/day initial doses showed a mean decrease in Time to Stand (TTSTAND) velocity (-0.011 ± 0.1248 [CI: -0.068, 0.046], n=21, p=0.6969) after 30-months that was not significantly different from VBP15-002 baseline **(Figure 2a)**
- Non-significant mean increases in Time to Climb 4 Stairs (TTCLIMB) velocity (0.035 ± 0.1733 [CI: -0.051, 0.121], n=18, p=0.4055), TTRW velocity (0.061 ± 0.6698 [CI: -0.272, 0.394], n=18, p = 0.7047), 6-Minute Walk Test (6MWT) total distance (32.0 ± 92.01 [CI: -18.954, 82.954], n=15 p=0.1994), and total North Star Ambulatory Assessment (NSAA) scores (1.6 ± 9.11 [CI: -2.921, 6.144], n=18, p=0.4635), were observed for the 2.0 and 6.0 mg/kg/day initial dose group after 30 months **(Figure 2b – 2e)**
- Mean increases in Quantitative Muscle Testing (QMT) knee flexion, knee extension, elbow extension, and elbow flexion scores were observed; however, interpretation of QMT results was limited by few observations at the Month 24 assessment

Figure 2. Efficacy of vamorolone on motor function assessments by high vs. low starting dose

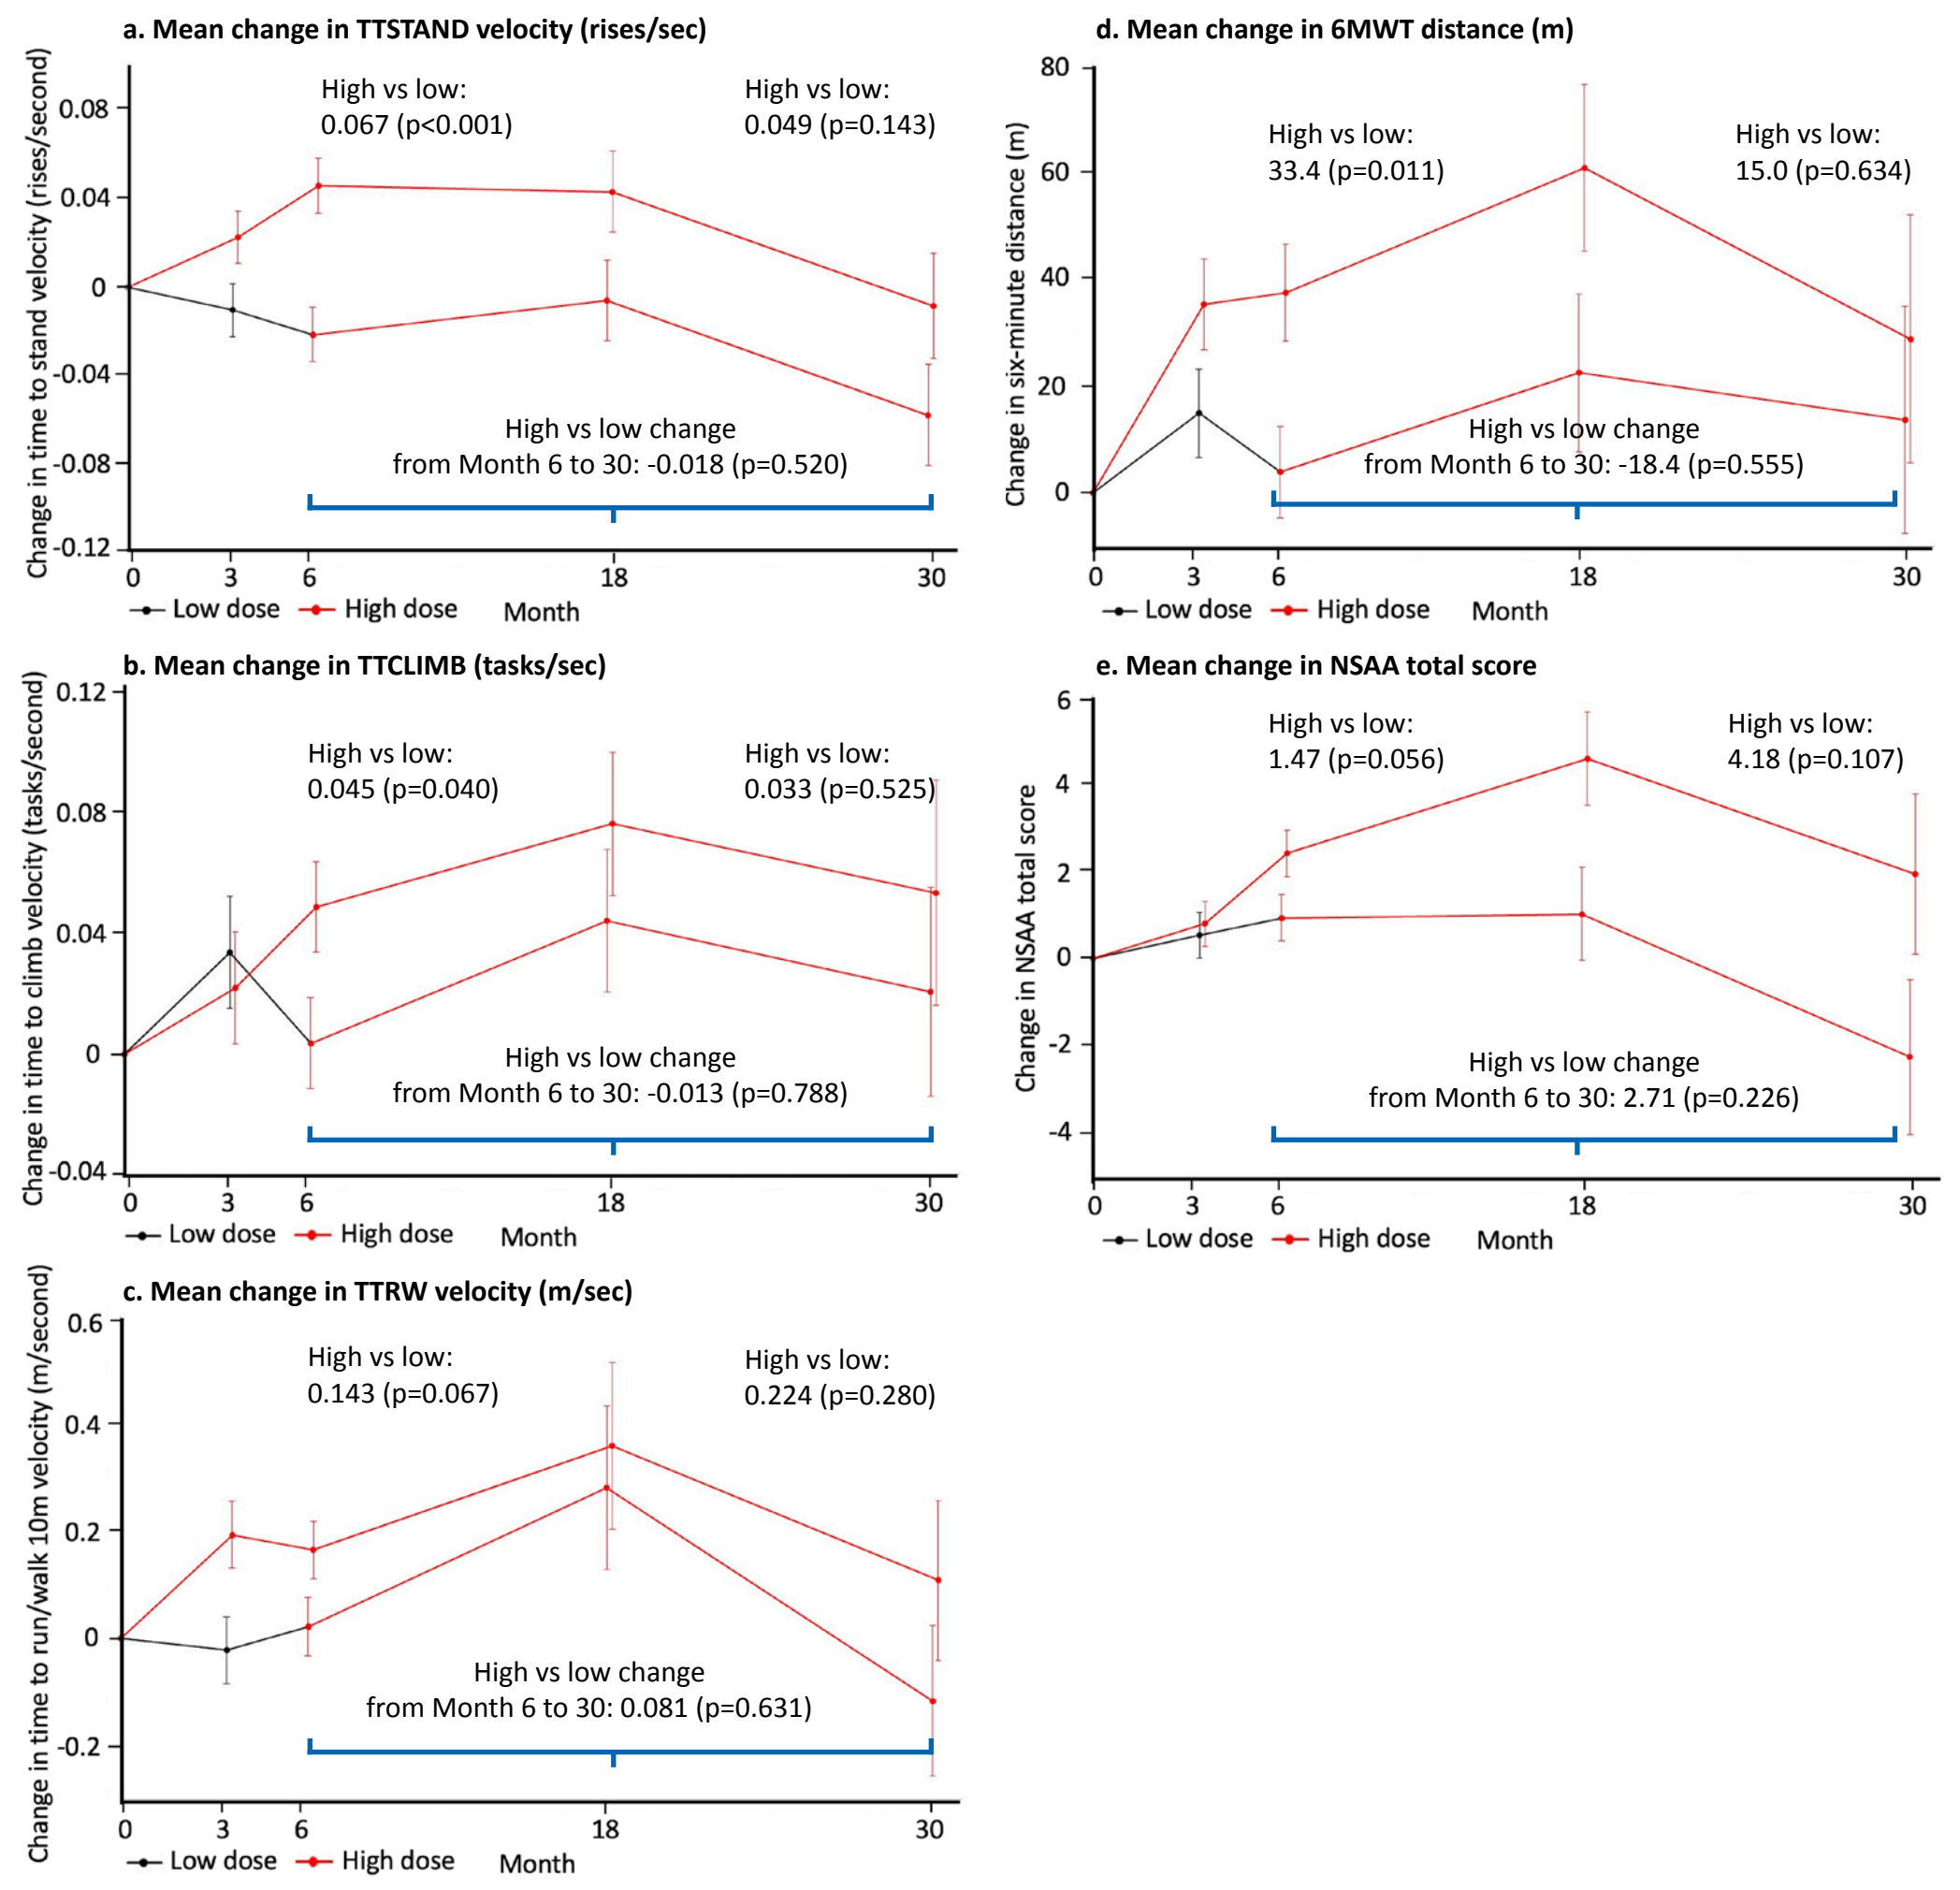

a. Mean change in TTSTAND velocity (rises/sec)
Change in time to stand velocity (rises/second)
High vs low: 0.067 (p<0.001)
High vs low: 0.049 (p=0.143)
High vs low change from Month 6 to 30: -0.018 (p=0.520)
b. Mean change in TTCLIMB (tasks/sec)
Change in time to climb velocity (tasks/second)
High vs low: 0.045 (p=0.040)
High vs low: 0.033 (p=0.525)
High vs low change from Month 6 to 30: -0.013 (p=0.788)
c. Mean change in TTRW velocity (m/sec)
Change in time to run/walk 10m velocity (m/second)
High vs low: 0.143 (p=0.067)
High vs low: 0.224 (p=0.280)
High vs low change from Month 6 to 30: 0.081 (p=0.631)
d. Mean change in 6MWT distance (m)
Change in six-minute distance (m)
High vs low: 33.4 (p=0.011)
High vs low: 15.0 (p=0.634)
High vs low change from Month 6 to 30: -18.4 (p=0.555)
e. Mean change in NSAA total score
Change in NSAA total score
High vs low: 1.47 (p=0.056)
High vs low: 4.18 (p=0.107)
High vs low change from Month 6 to 30: 2.71 (p=0.226)
Low dose
High dose
Month

Safety and tolerability

- During the 24-month VBP15-LTE Treatment Period, 46 participants (100%) experienced at least one treatment-emergent AE (TEAE)
- The most common TEAEs are shown in **Table 1**
- Ten subjects de-escalated the vamorolone dose from 6.0 mg/kg/day to 2.0 mg/kg/day due to a TEAE of weight increase
- According to local site AE reporting, 5/39 (12.8%) patients were observed to have a total of 6 clinical fracture events
- Mean changes in hematology, chemistry, lipids, or urine analysis parameters were minimal and clinically unremarkable
- Mean values for glutamate dehydrogenase and gamma-glutamyl transferase were normal at all VBP15-LTE timepoints

Table 1. AEs by dose at time of event

	Vamorolone dose at the time of the event (mg/kg/day)				
Overall	0.25 mg/kg (N = 11)	0.75 mg/kg (N = 23)	2.0 mg/kg (N = 38)	4.0 mg/kg (N = 3)	6.0 mg/kg (N = 41)
Any TEAE	4 (36.4)	14 (60.9)	29 (76.3)	1 (33.3)	39 (95.1)
Any treatment-related TEAE	0	0	8 (21.1)	1 (33.3)	23 (56.1)
Any TEAE with CTCAE Grade ≥ 3	0	1 (4.3)	0	0	1 (2.4)
Any TEAE leading to Discontinuation of Study	0	0	1 (2.6)	0	0
Any SAE*	0	1 (4.3)	0	0	1 (2.4)
Any Serious TEAE	0	1 (4.3)	0	0	1 (2.4)
Preferred Term					
Nasopharyngitis	0	1 (4.3)	7 (18.4)	0	12 (29.3)
Cough	1 (9.1)	1 (4.3)	7 (18.4)	0	8 (19.5)
Pyrexia	0	0	9 (23.7)	0	8 (19.5)
Vomiting	1 (9.1)	0	5 (13.2)	1 (33.3)	7 (17.1)
Weight increased	0	0	2 (5.3)	0	10 (24.4)
Constipation	0	0	3 (7.9)	0	8 (19.5)
Pain in extremity	0	3 (13.0)	5 (13.2)	0	6 (14.6)
Fall	0	1 (4.3)	4 (10.5)	0	6 (14.6)
Upper respiratory tract infection	0	2 (8.7)	5 (13.2)	0	6 (14.6)
Headache	0	0	4 (10.5)	0	7 (17.1)
Influenza	0	1 (4.3)	3 (7.9)	0	6 (14.6)

*No TEAEs led to discontinutation of study drug. No deaths occurred.
CTCAE, Common Terminology Criteria for Adverse Events; SAE, Serious adverse events.

- There was no significant difference in changes to mean Body Mass Index (BMI) z-scores between the vamorolone and Duchenne Natural History Study (DNHS) participants
- Comparison of vamorolone- to GC-treated DNHS participants showed a significant difference (p=8.94 ×10-07) in mean height percentile change, with no evidence of growth deceleration in the vamorolone group

Conclusions

- Increase in TTSTAND velocity in vamorolone-treated DMD boys at younger ages followed by a decrease in TTSTAND velocity as the boys get older is consistent with longitudinal observations for timed function tests, including TTSTAND, in GC-treated boys with DMD
- Motor function improved from baseline to month 18 and returned to baseline by month 30, when natural history suggests motor function would usually have declined to below baseline by this time; in keeping with observations with other steroids, natural history was therefore modified with vamorolone (a 2.5 year delay in decline in function)
- Modest functional improvements were observed in TTCLIMB velocity, TTRW velocity, NSAA total score, and 6MWT distance for participants on 2.0 and 6.0 mg/kg/day initial dose over a 30-month treatment period
- The mean increase in 6MWT total distance walked showed a clinically meaningful[5] vamorolone treatment effect of >30 meters from baseline (VBP15-002) to Month 24 (VBP15-LTE)
- Continuous treatment for up to 30 months was not associated with linear growth deceleration
- Mean change in BMI z-score was not significantly different with vamorolone compared to GC-treated DNHS participants
- Over a 30-month treatment period, at doses of up to 6.0 mg/kg/day, vamorolone appears to be well-tolerated with 89.1% of patients completing the study, and only 1 TEAE leading to discontinuation, and 2 SAE or serious TEAEs, were observed
- In addition, 10 subjects (24%) down titrated due to weight gain but remained on treatment, and there was no impact on linear growth trajectory observed

References

1. Heier CR, et al. *Life Sci Alliance*. 2019;2(1):e201800186; 2. Conklin LS, et al. *Pharmacol Res*. 2018; 136:140-150; 3. Hoffman EP, et al. *Neurology*. 2019; 93(13): e1312-e1323; 4. Smith EC, et al. *PLoS Med*. 2020;17(9):1–18; 5. Henricson E, et al. *PLoS Curr*. 2013;5.

Acknowledgements