Pediatric Leber's Hereditary Optic Neuropathy (LHON): Real-world efficacy results following long term idebenone treatment

Xavier Llòria
Santhera Pharmaceuticals (Switzerland) Ltd, Pratteln, Switzerland

Magda Silva
Santhera Pharmaceuticals (Switzerland) Ltd, Pratteln, Switzerland

Claudia Catarino
Friedrich-Baur-Institute, Munich, Germany

Guenther Rudolph
University Hospital of the Ludwig-Maximilians-Universität, Munich, Germany

Felice Lob
University Hospital of the Ludwig-Maximilians-Universität, Munich, Germany

Bettina von Livonius
University Hospital of the Ludwig-Maximilians-Universität, Munich, Germany

Thomas Klopstock
Friedrich-Baur-Institute, Munich, Germany

Poster presented at the 14th EUNOS, June 16–19, 2019. Porto, Portugal. Poster Session I. Poster PO54

Background

- LHON results in bilateral, severe central vision loss, and is caused by mitochondrial DNA mutations
- LHON is typically diagnosed between 15 – 30 years of age, although it can be detected earlier
- Idebenone, the only approved treatment for LHON in Europe*[1] has been shown to be efficacious and safe in a large proportion of adult patients, but pediatric data is limited
- Priglinger C. et al.[2] observed good safety in 32 LHON patients between 4 and 16 years of age, treated with idebenone (300 mg three times a day) under real-world conditions. Patients below the age of 13 seemed to have better efficacy than patients between 13 and 16 years of age. Erdei Z. et al. observed improvement of visual acuity (VA) and visual fields in a 10 year old female treated over 12 months with idebenone (900 mg/day)[3]

Objectives

To investigate the safety and benefit of long-term idebenone treatment on the VA in pediatric patients treated under routine clinical practice in an Expanded Access Program (EAP)[4]

Methods

- Retrospective medical chart analysis of patients who were under 12 years of age at start of therapy
- All patients received idebenone 900 mg (Raxone® 150 mg tablets) in 3 divided doses through a Named Patient Scheme under routine clinical practice
- VA expressed as logMAR; all Snellen converted VA > 1.68 logMAR and off-chart values are imputed to 1.8 logMAR
- Safety measured through frequency and severity of adverse events

Results

Demographics and Baseline

- Five pediatric patients, 2 female and 3 male, age between 6.9 and 11 years at baseline (BL; median age 9.8 years)
- Time since onset ranged from 1.7 months to 5 years
- Patients carried one of the following mutations, G3460A *(Figure 1)* T14484C (Figure 2) G11778A (Figure 1) A14495G *(Figure 1)*
- Best VA at BL ranged between 0.16 to 1.20 logMAR (median 0.94 logMAR)
- For all patients, Nadir occured at BL for at least one eye

Time in Treatment

- Median time in treatment was 33.64 months, range between 6.8 to 40 months

Change in Visual Acuity

- Median best VA at last visit (LV) was 0.08 logMAR (range -0.18 to 1.36)
- Magnitude of recovery 2 to 9 ETDRS lines at first observation of clinically relevant recovery (CRR), which increased to 4 to 12 ETDRS lines by LV

Safety

- No new safety signals were observed

Discussion

Patient 1

The most recent eye deteriorated to a Nadir despite treatment with idebenone for nearly 4 months. With maintained treatment both eyes improved although at a different rate and with different magnitude

Patient 2

Patient showed an initial (erratic) improvement over the first 12 months but deteriorated (despite therapy) back to BL levels over the next 2 years. VA never deteriorated to off-chart

Patient 3

This patient showed a positive response after the first 3 months of therapy which normalized the VA after 25 months of treatment

Patient 4

A rare mutation carrier with a moderate visual function loss and a fast and progressive response, resulting in normalization of VA after approximately 3 months on treatment

Patient 5

Treatment was started after 4 years since disease onset. Visual deterioration was very asymmetric and VA in the worst eye showed a temporary improvement (artefact?). Overall, VA was maintained at initial levels

Dose in LHON pediatric population

The safety and efficacy of Raxone in LHON patients under 12 years of age have not yet been established[1]

Pharmacokinetic data from population pharmacokinetic studies, which included pediatric Friedreich's Ataxia patients of age 8 years and above, did not reveal any significant differences in the pharmacokinetics of idebenone. In clinical trials in Friedreich's Ataxia, 32 patients between the ages of 8 and 11 years and 91 patients between the ages of 12 and 17 years received idebenone at ≥ 900 mg/day for up to 42 months[1]

The data presented here indicates that the safety profile of idebenone is not different from that observed in adolescents and adults and VA in pediatric patients as young as 7 years of age can improve/prevent deterioration to off-chart VA with idebenone

Patient eyes VA along treatment

Figure 1

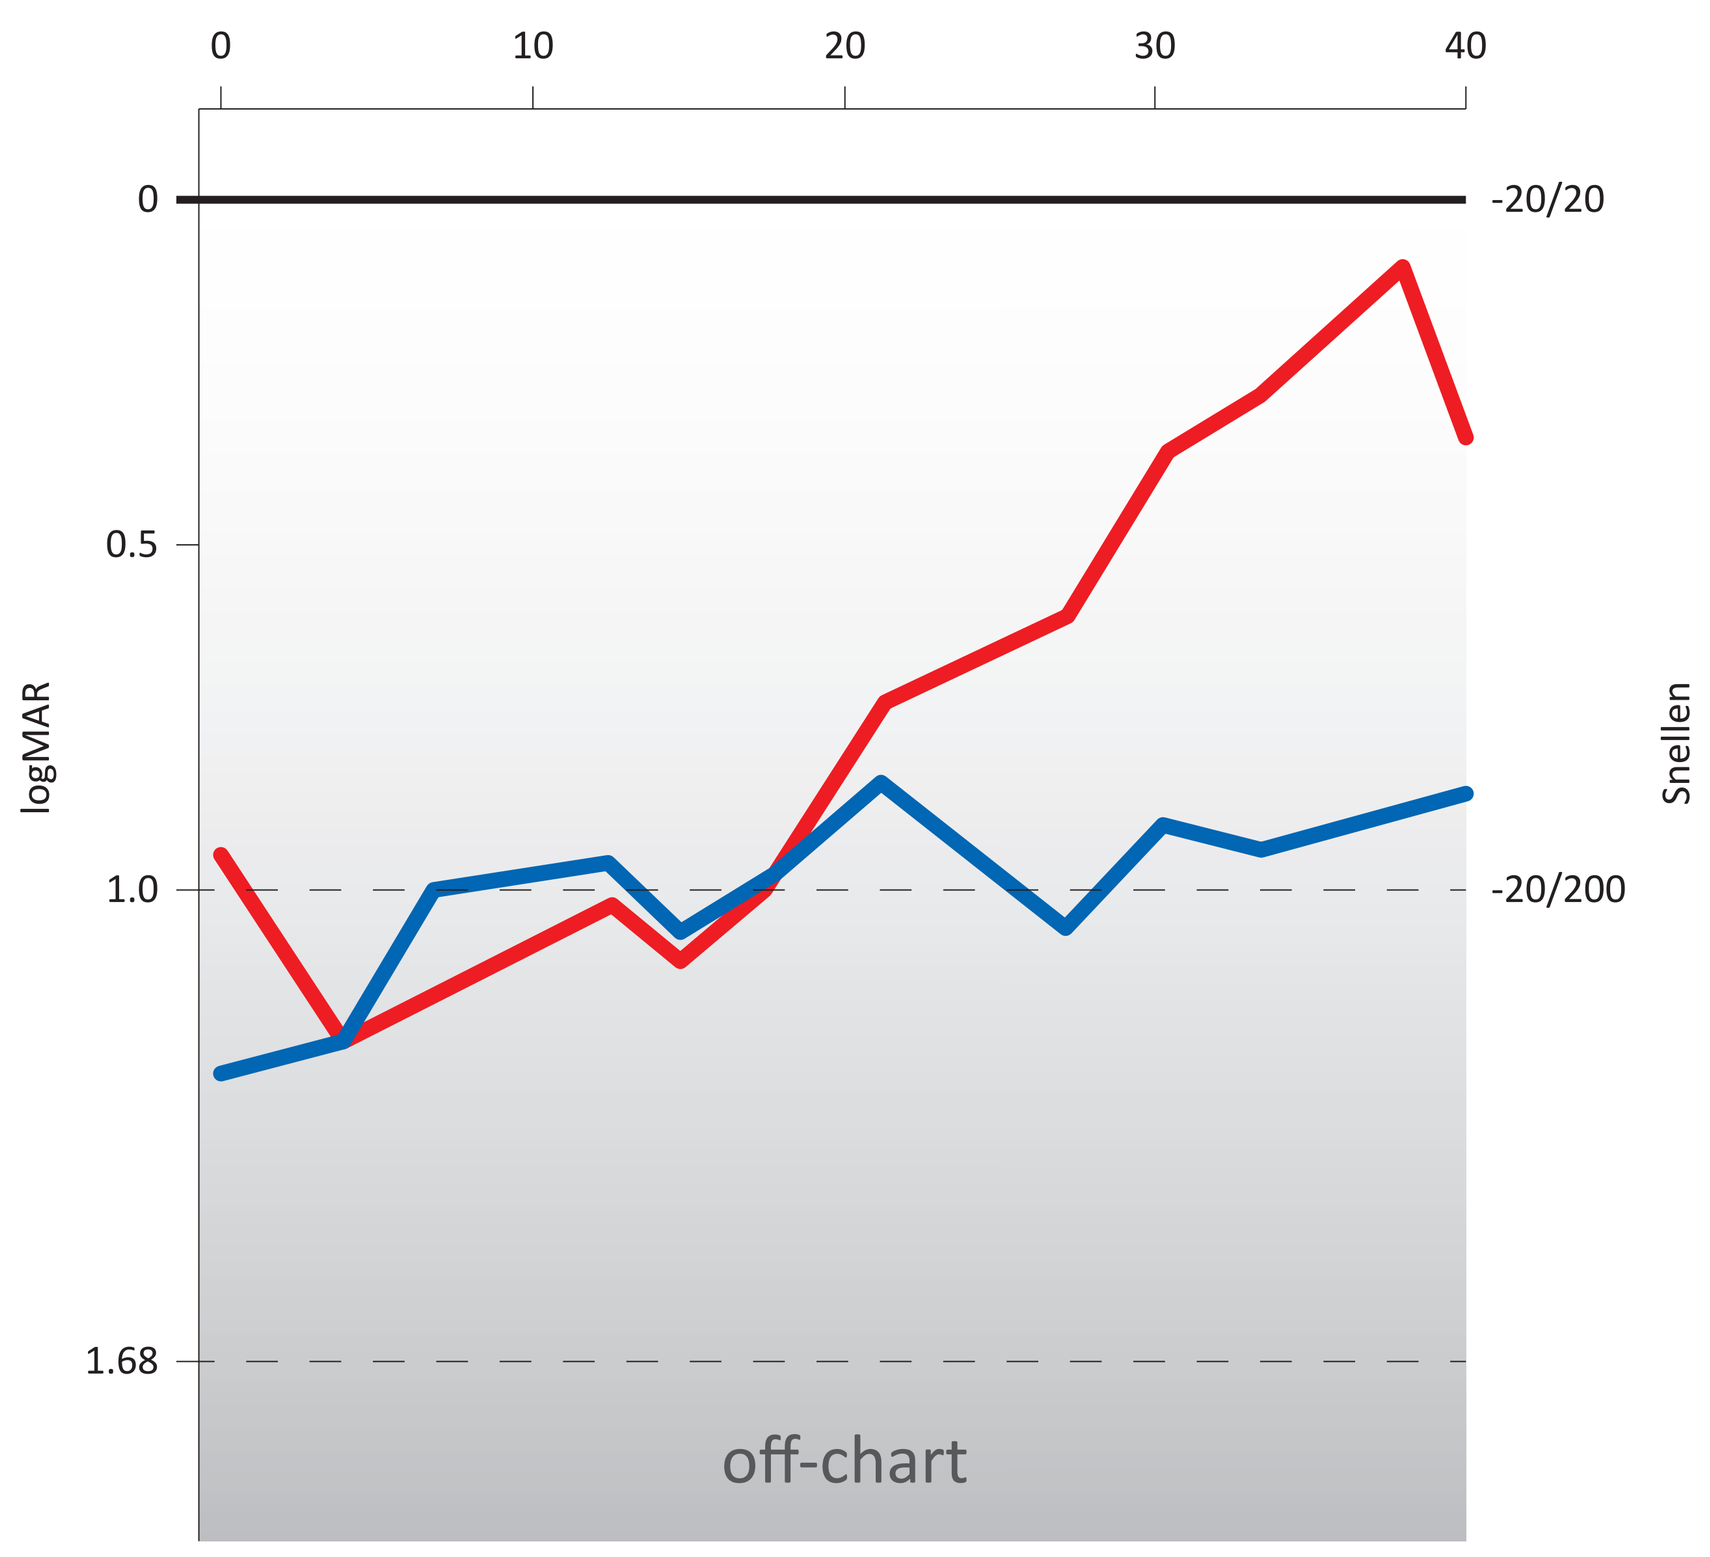

Months in Treatment
0
10
20
30
40
0
0.5
1.0
1.68
logMAR
Snellen
-20/20
-20/200
off-chart

G3460A carrier, 6.9 years at BL

Left	Right
1.7 months onset at BL 0.94 logMAR at BL Nadir 3.9 months post BL Gain of 6 lines from BL Gain of 8 lines and 4 letters from Nadir	3.5 months onset at BL 1.26 logMAR at BL Nadir at BL Gain of 4 lines from BL/Nadir

Figure 1

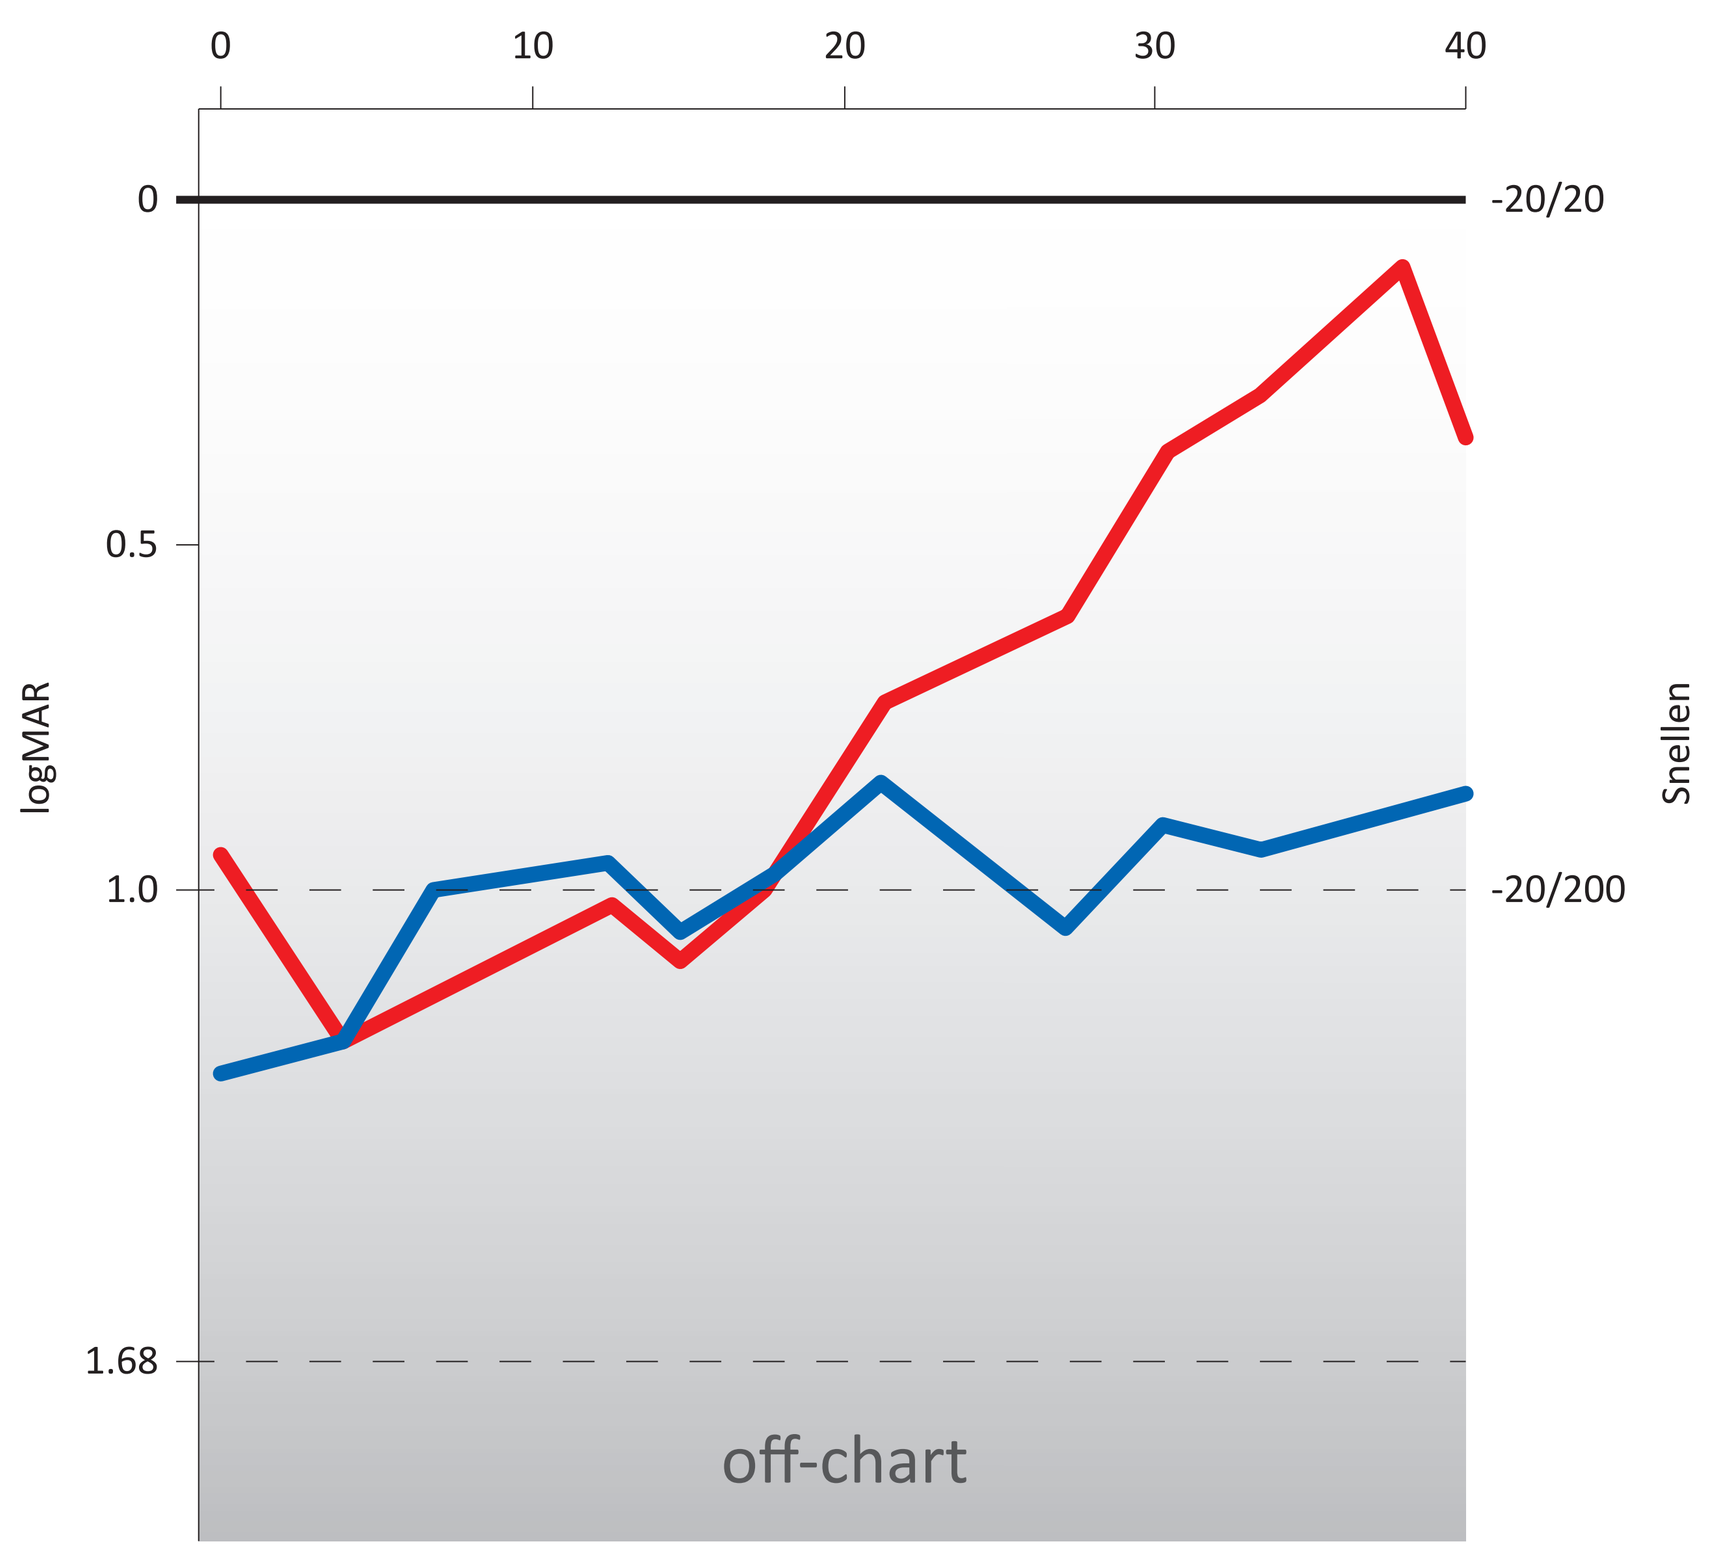

Months in Treatment
0
10
20
30
40
0
0.5
1.0
1.68
logMAR
Snellen
-20/20
-20/200
off-chart

T14484C carrier, 9.1 years at BL

Left	Right
7.9 months onset at BL 1.42 logMAR at BL Nadir at BL Gain of 1 letter from BL/Nadir	7.9 months onset at BL 1.20 logMAR at BL Nadir at LV Loss of 1 line and 3 letters from BL

Figure 1

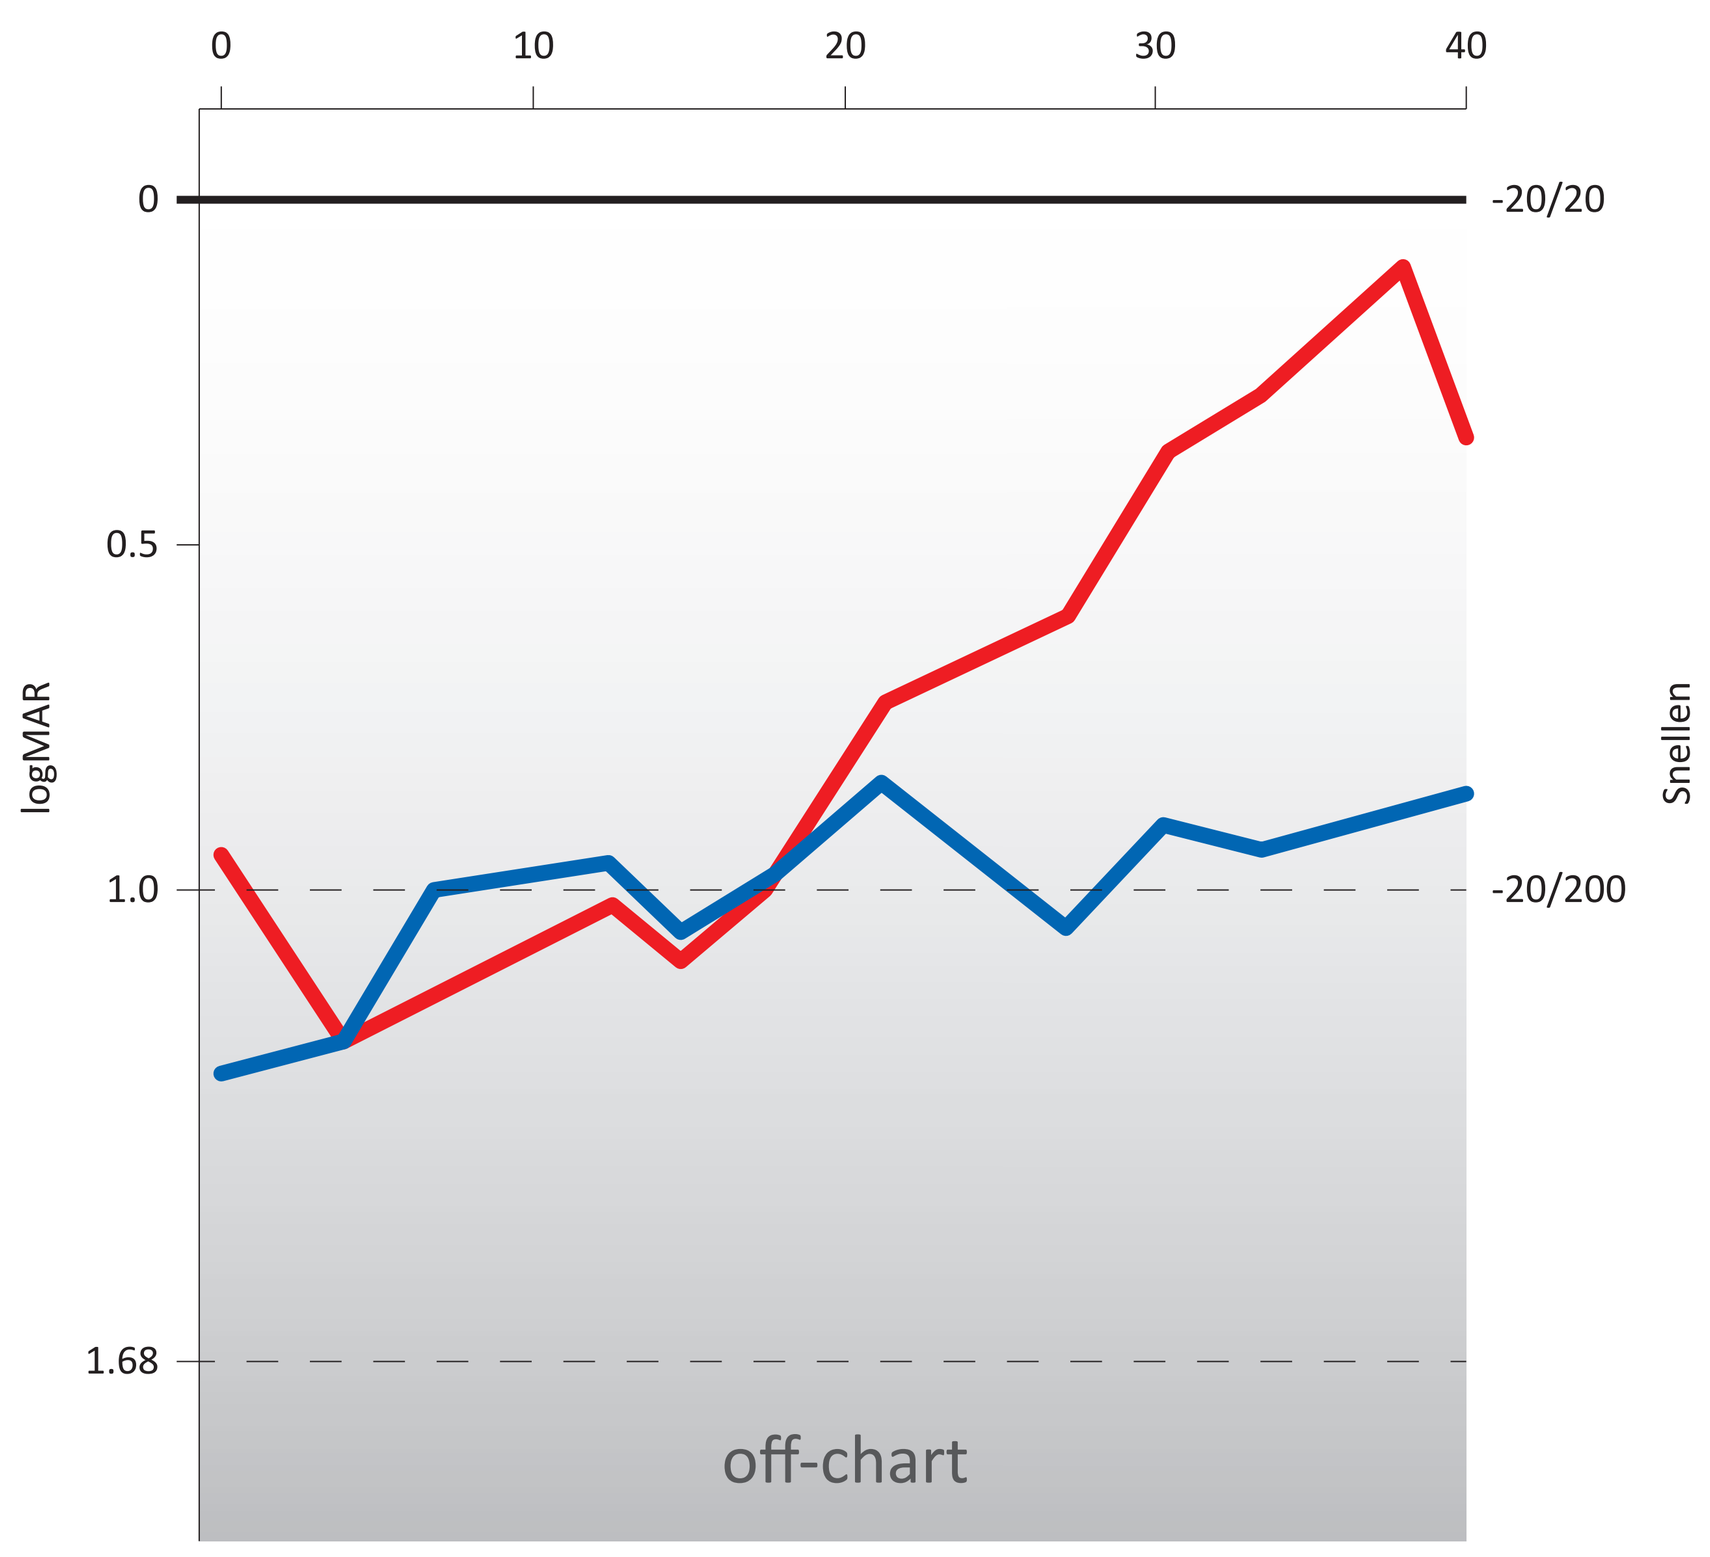

Months in Treatment
0
10
20
30
40
0
0.5
1.0
1.68
logMAR
Snellen
-20/20
-20/200
off-chart

T14484C carrier, 9.8 years at BL

Left	Right
6.0 months onset at BL 1.18 logMAR at BL Nadir at BL Gain of 12 lines and 3 letters from BL/Nadir	6.0 months onset at BL 1.06 logMAR at BL Nadir at BL Gain of 10 lines and 4 letters from BL/Nadir

Figure 4

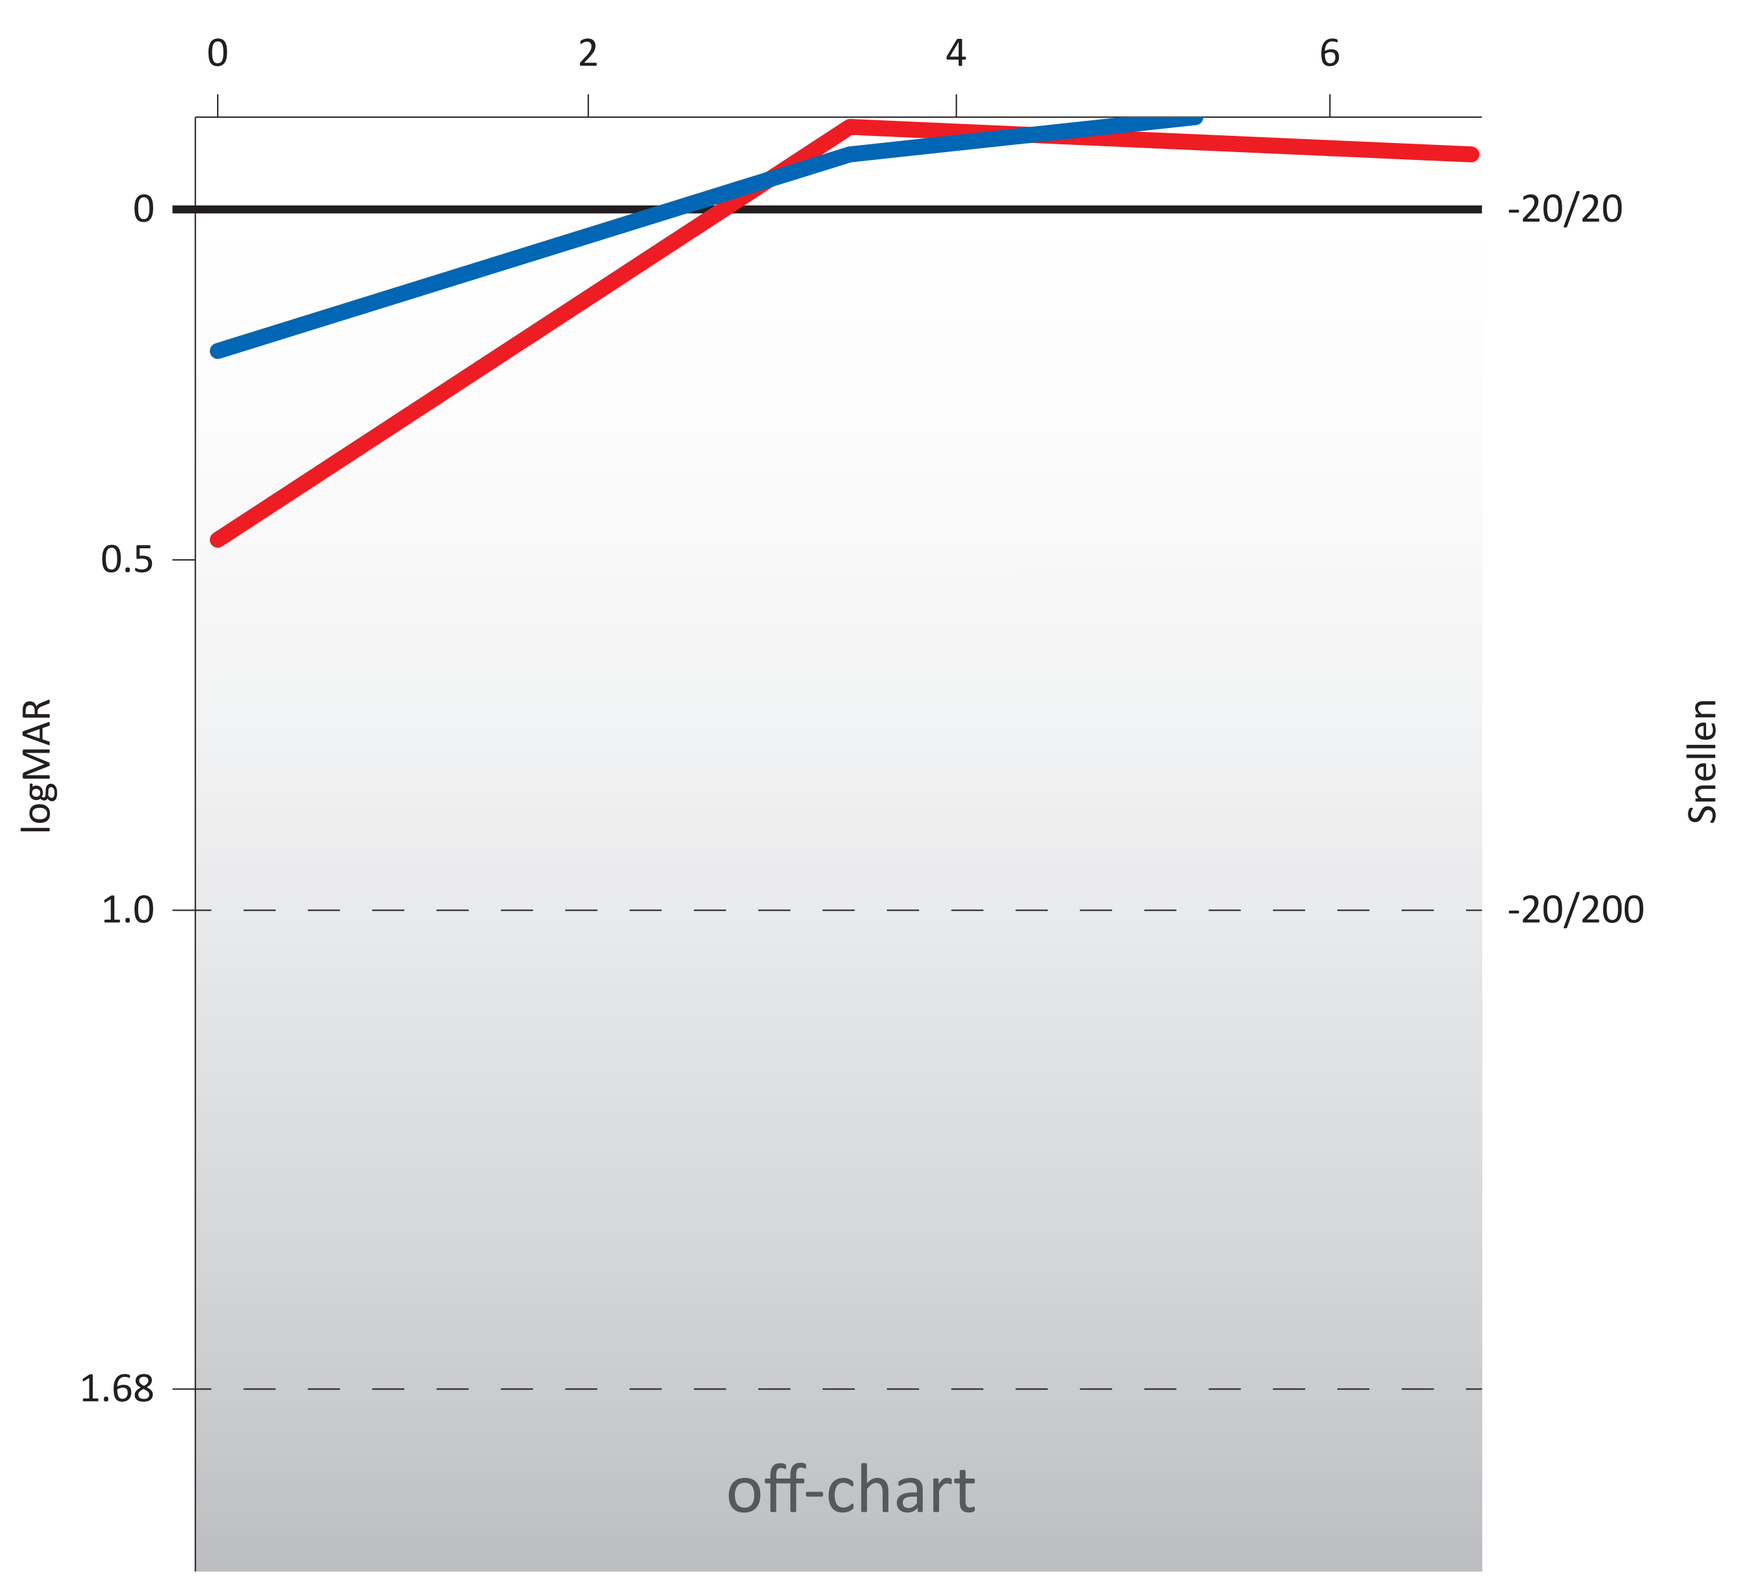

Months in Treatment
0
2
4
6
0
0.5
1.0
1.68
logMAR
Snellen
-20/20
-20/200
off-chart

A14495G carrier, 11 years at BL

Left	Right
2.4 months onset at BL 0.46 logMAR at BL Nadir at BL Gain of 5 lines and 2 letters from BL/Nadir	1.7 months onset at BL 0.20 logMAR at BL Nadir at BL Gain of 3 lines and 4 letters from BL/Nadir

Figure 5

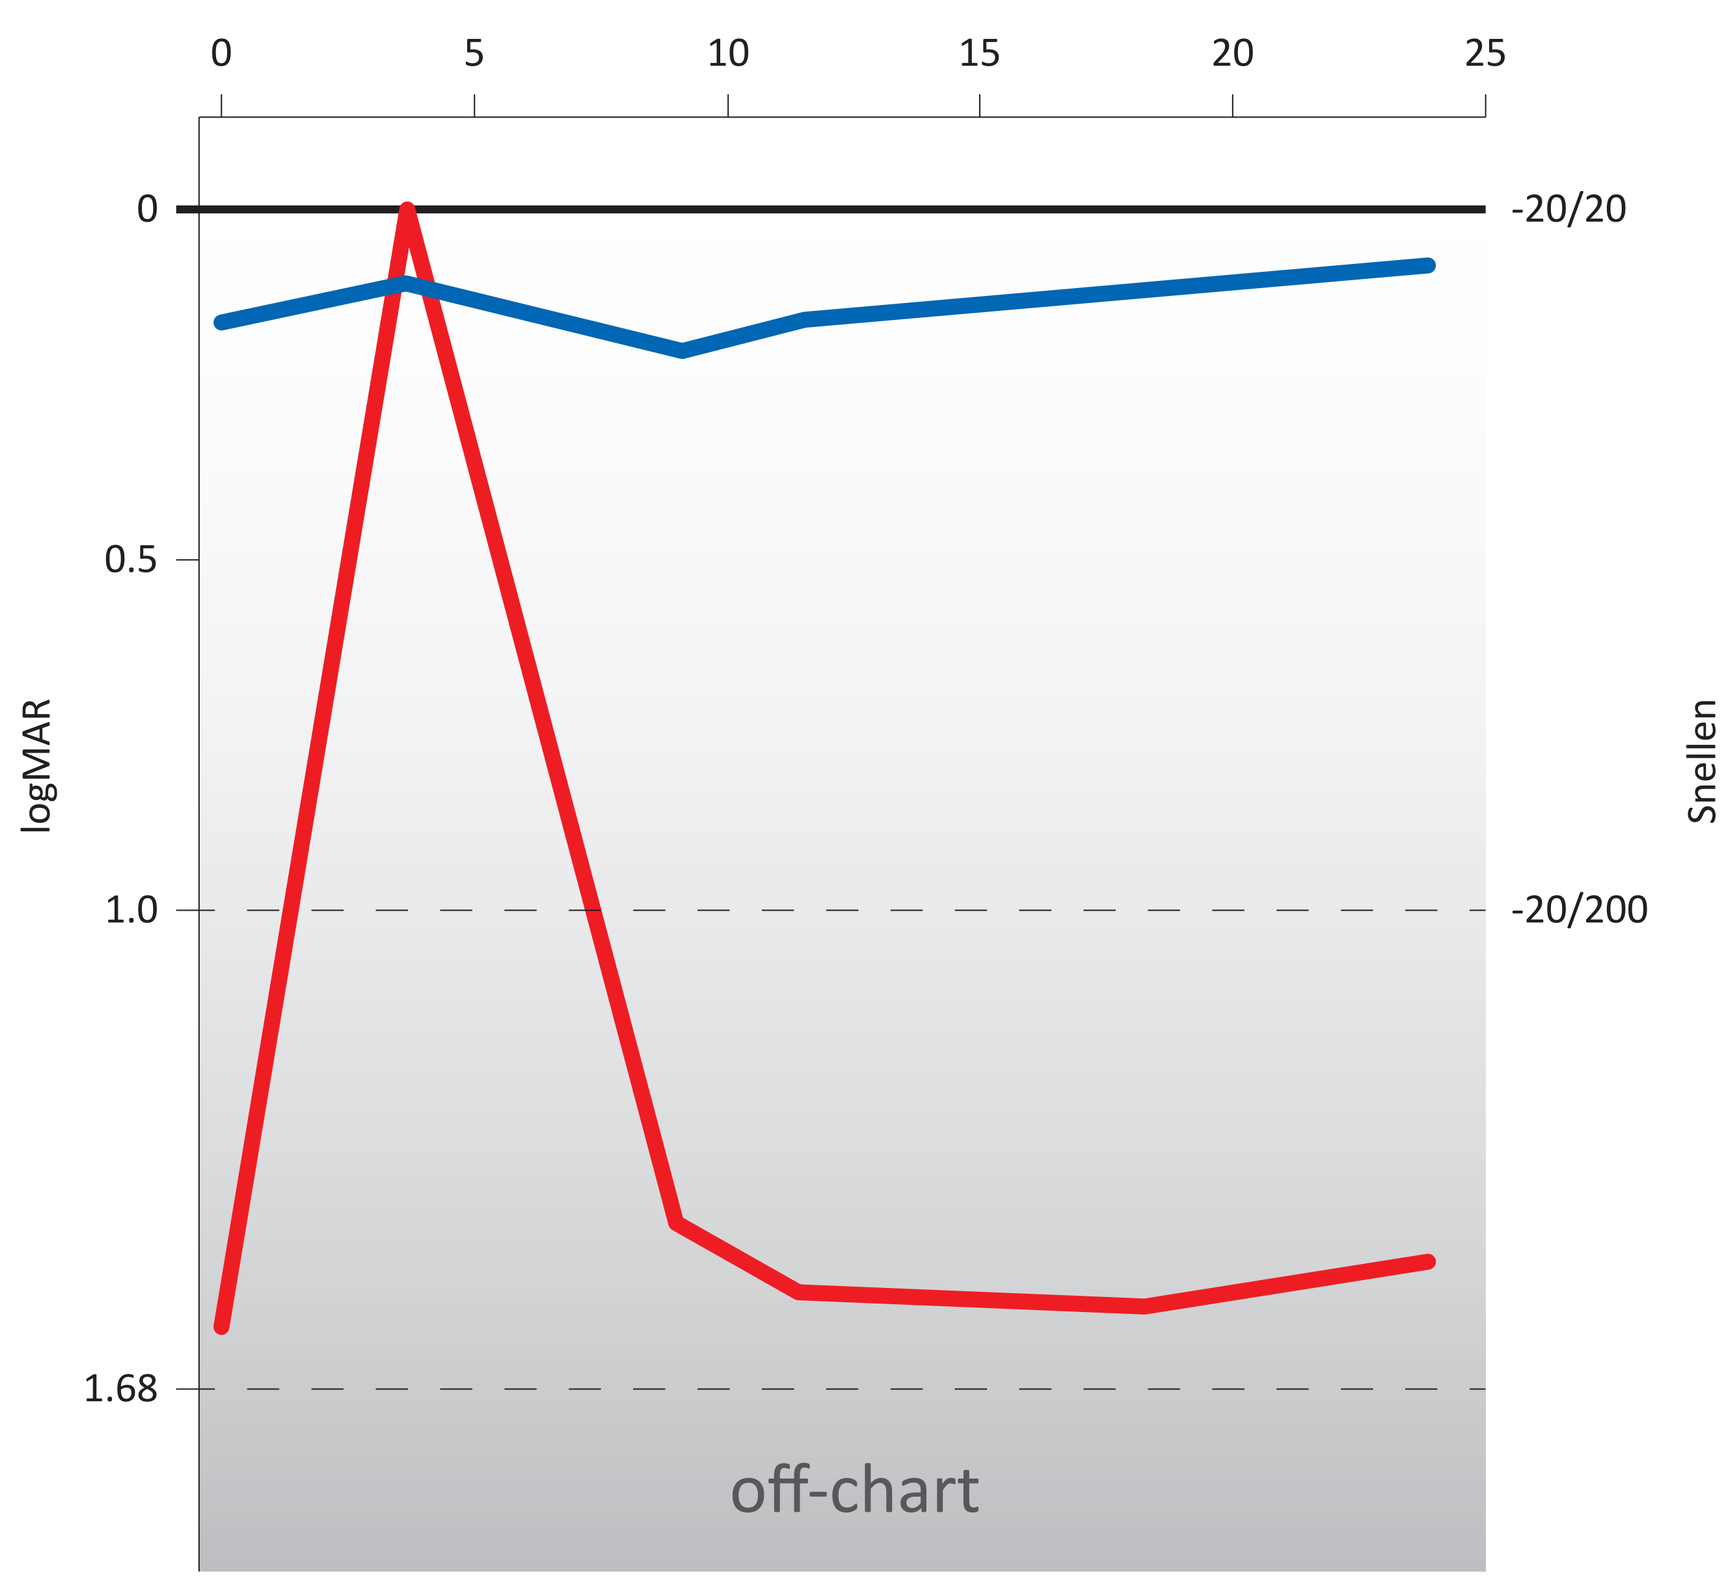

Months in Treatment
0
5
10
15
20
25
0
0.5
1.0
1.68
logMAR
Snellen
-20/20
-20/200
off-chart

G11778A carrier, 11 years at BL

Left	Right
4.1 years onset at BL 1.58 logMAR at BL Nadir at BL Gain of 4 letters from BL/Nadir	4.1 years onset at BL 0.16 logMAR at BL Nadir 9 months post BL Gain of 4 letters from BL Gain of 1 line and 1 letter from BL/Nadir

Conclusions

Results of this real-world program with idebenone in LHON pediatric patients suggest:

- No additional safety signals specific to paediatric population were reported from use in this small number of patients below age 12†
- Treatment with idebenone can result in both stabilization of good residual VA and relevant recovery of lost vision in some patients
- In some eyes/subjects the beneficial effect can continue to progressively improve even after 30 months of therapy. None of the eyes treated deteriorated to off-chart VA over the observation period
- These data indicate that long-term idebenone treatment could be a safe and efficacious therapeutic approach in pediatric LHON patients

Acknowledgements

The authors would like to thank all patients and health care professionals participating in this Expanded Access Program for their contribution in collecting the data. If you have any questions about this data please contact Dr Xavier Llòria (Xavier.Lloria@Santhera.com).

Conflict of interest

Xavier Llòria and Magda Silva are regular employees of Santhera Pharmaceuticals (Switzerland) Ltd. Thomas Klopstock has been investigator in Santhera sponsored trials, has served on the Scientific Advisory Board and received speaker honoraria from Santhera. Felice Lob and Claudia Catarino have received speaker honoraria from Santhera.

References

1. Raxone® Summary of Product Characteristics (SmPC). European Medicines Agency, 2018
2. Priglinger C. et al. A retrospective analysis of childhood onset Leber's Hereditary Optic Neuropathy (LHON) treated with idebenone Poster Presentation. European Pediatric Ophthalmological Society (EPOS). Budapest, Hungary. 7-8 September 2018
3. Erdei Z. et al. Clinical experience with idebenone in the treatment of adolescents with Leber's Hereditary Optic Neuropathy – 3 case reports. Poster Presentation. European Pediatric Ophthalmological Society (EPOS). Budapest, Hungary. 7-8 September 2018
4. Raxone® EPAR, European Medicines Agency, September 2015

† Insufficient data from controlled studies still exist with Raxone in LHON paediatric patients to date.

Raxone®, Santhera Pharmaceuticals (Deutschland) GmbH, idebenone 150 mg tablets

* In the European Union, Raxone® is indicated for the treatment of visual impairment in adolescent and adult patients with Leber's Hereditary Optic Neuropathy (LHON)

Job code: NP-HQ-LHON-RAX-0004

santhera
THEIR FUTURE – OUR FOCUS

06_SANTH_05_19_004_Pediatric Nanos Poster_ns.indd 1
17/06/2019 08:39