Crossing thresholds and changing rates of respiratory function decline are predictive of clinical outcomes in Duchenne Muscular Dystrophy (DMD)

Craig McDonald[1], Heather Gordish-Dressman[2], Mika Leinonen[3], Oscar H Mayer[1] and CINRG-DNHS Investigators

[1]UC Davis Health, Davis, USA, [2]The George Washington University School of Medicine and Health Sciences, Washington DC, USA, [3]Clinical Data Science, Basel, Switzerland, [1]Children's Hospital of Philadelphia (CHOP), Philadelphia, USA

Poster presented at MDA April 13th – 17th, 2019; Poster: 11

Background

- Preservation of absolute FVC and FVC%p earlier in adolescence is associated with delayed pulmonary function decline,[1] with decreased risk of pulmonary complications including the need for assisted ventilation[2,3] and crossing of the FVC <1L threshold – associated with a 4-fold increased risk of death.[1,4]
- A prior study reported that falling below FVC of 1L was associated with a 5-year survival of only 8%.[4] However, with recent improvements in standards of care, these findings are now outdated.
- Current standards of care[3] are based on expert opinion rather than actual pulmonary data and utilize FVC%p thresholds to help guide clinical management, including the use of cough assist and nocturnal and diurnal ventilation.[1]
- No systematic analyses have assessed the impact of crossing thresholds or changing rate of respiratory function decline in delaying time to reaching clinically relevant outcomes.
- Here we present analyses from the Cooperative International Researchers Group Duchenne Natural History Study (CINRG-DNHS)[1,5] to assess the predictive value of crossing clinically relevant thresholds and different rates of decline on the risk of assisted ventilation and death.

Objectives

To conduct a systematic analysis of data from CINRG-DNHS to assess:

- If falling below a 1L threshold in FVC is still predictive of 5-year survival or assisted ventilation, or if any other threshold of FVC/FVC%p or PEF/PEF%p predicts the 5-year survival or assisted ventilation.
- If rate of decline in FVC%p or PEF%p is predictive of the time to death and the initiation of assisted ventilation.

Methods

Analysis 1: Crossing thresholds of FVC or PEF

- Conducted in patients with at least one assessment at or above and one below the FVC/PEF threshold of interest. The time when the threshold was crossed is used as the baseline for the analysis.
- Thresholds were scanned in small decrements until <5 events occurred. Kaplan-Meier method was used to derive 5-year and 2-year risk estimates (and 95% CI) of death and assisted ventilation.
- Similar analyses were conducted for FVC%p and PEF%p.

Analysis 2: Changes in rate of decline of FVC%p or PEF%p

- Conducted in patients with at least one assessment of FVC%p or PEF%p between 80 – 50% during the study. All values between 80 – 30% were used for each patient to estimate individual annual rates of decline.
- The individual rates of decline were categorized using a cut-off of 5%, the observed mean in natural history studies, as those with a more rapid decline rate (>5% per year) or less rapid rate (<5% per year). Sensitivity analyses were conducted with cut-offs at 4%, 6% and 7%.
- Time from crossing 50% of FVC%p or PEF%p for the first time was used as the baseline for the analysis and time to death or start of ventilation was estimated with Kaplan-Meier method. Sensitivity analyses were conducted for time from crossing 45%, 55% and 60% thresholds.
- The two groups (those with a more rapid rate of decline versus those with a less rapid rate) were compared with Kaplan-Maier methods. P-values were calculated with the log-rank test.

Figure 1. CINRG-DNHS analysis data set

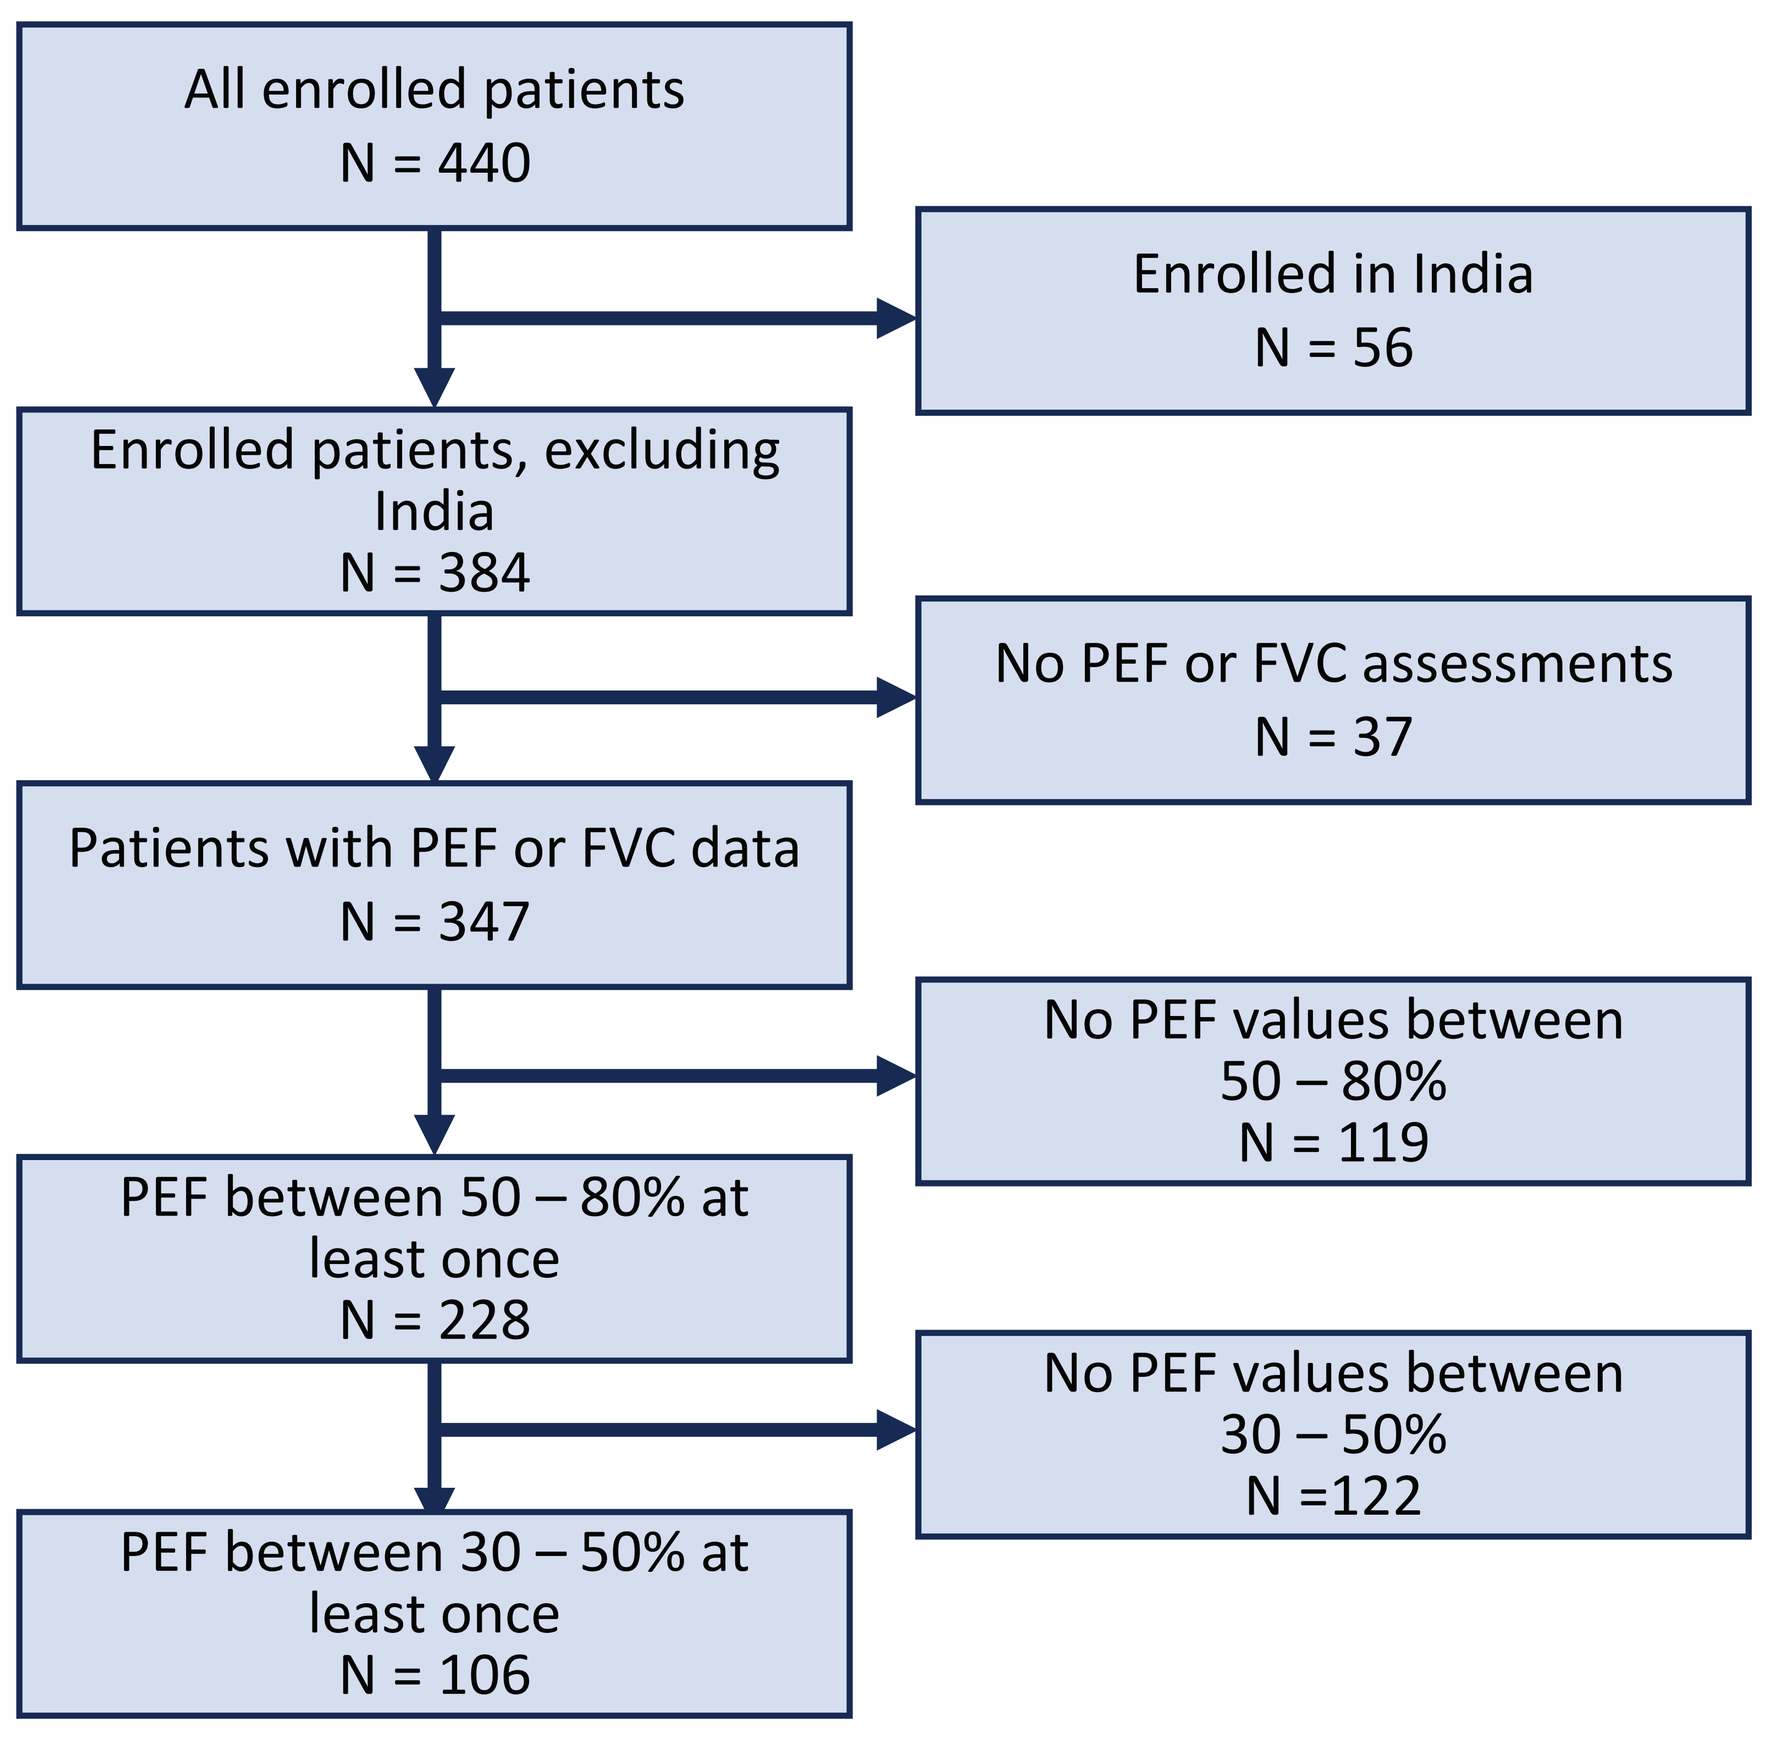
All enrolled patients
N = 440
Enrolled in India
N = 56
Enrolled patients, excluding India
N = 384
No PEF or FVC assessments
N = 37
Patients with PEF or FVC data
N = 347
No PEF values between 50 – 80%
N = 119
PEF between 50 – 80% at least once
N = 228
No PEF values between 30 – 50%
N =122
PEF between 30 – 50% at least once
N = 106

Table 1. Disease characteristics for patients declining <5% versus ≥5% per year

	FVC%p Decline Group		PEF%p Decline Group	
	<5% / year (N = 29)	≥5% / year (N = 45)	<5% / year (N = 61)	≥5% / year (N = 45)
Age (at crossing 50%), years				
Mean (SD)	16.7 (3.0)	15.5 (1.9)	14.2 (4.2)	14.4 (3.2)
Median	16.8	15.3	14.2	15.0
Min, Max	11, 22	12, 19	6, 25	7, 20
Steroid use (at crossing 50%), n (%)				
Steroid-naïve	2 (6.9)	6 (13.3)	7 (11.5)	3 (6.7)
Ex-user	3 (10.3)	15 (33.3)	7 (11.5)	12 (26.7)
Current user	24 (82.8)	24 (53.3)	47 (77.0)	30 (66.7)
Ambulatory status (at crossing 50%), n (%)				
Ambulatory	7 (24.1)	2 (4.4)	28 (45.9)	6 (13.3)
Non-ambulatory	22 (75.9)	43 (95.6)	33 (54.1)	39 (86.7)
Peak PEF (L/min) during study				
Mean (SD)	272.3 (85.2)	244.4 (47.2)	263.4 (65.8)	228.0 (59.8)
Median	258.0	241.8	263.4	226.8
Min, Max	146, 533	172, 366	115, 533	84, 377
Peak FVC (L) during study				
Mean (SD)	2.14 (0.65)	2.03 (0.44)	2.23 (0.58)	1.95 (0.57)
Median	2.0	2.0	2.1	1.9
Min, Max	1.3, 4.7	1.2, 3.1	1.3, 4.7	0.9, 3.4

- 440 patients from US, Europe, Australia and India are included in the CINRG-DNHS, 56 patients from India were excluded from this analysis because of the possibility of differing standards of care. In addition, 37 patients with no PEF or FVC data were excluded from all analyses.
- The remaining patients were used to perform the analyses as applicable (**Figure 1**).
- Baseline characteristics for patients with annual FVC%p and PEF%p decline <5% versus ≥5% are shown in **Table 1**.

Results

Figure 2. 5-year survival estimates as a function of FVC (A) and PEF (B) thresholds and 2-year risk of initiation of assisted ventilation as a function of FVC%p (C) and PEF%p (D).

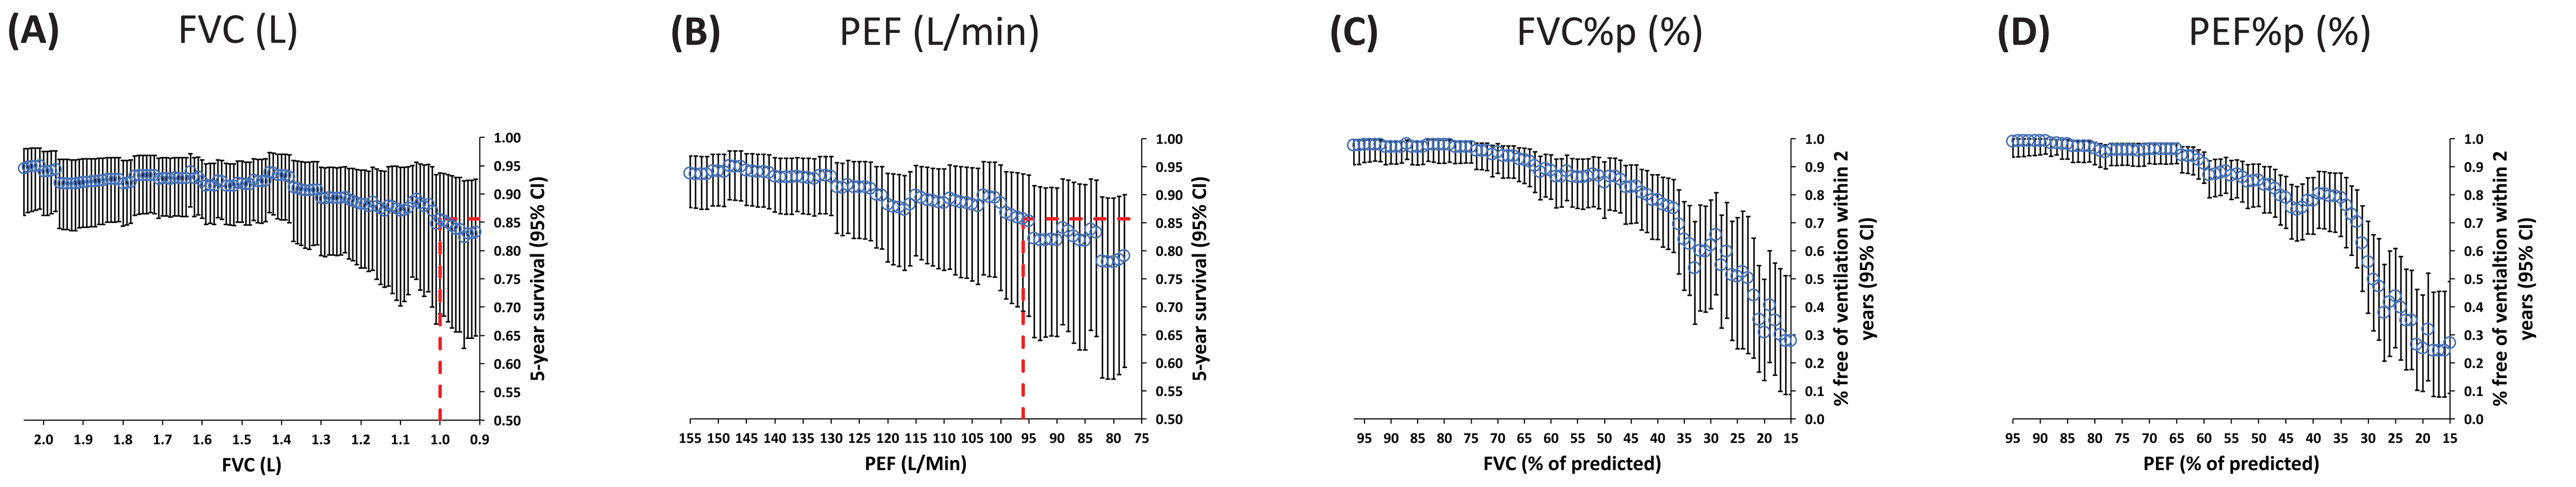
(A) FVC (L)
(B) PEF (L/min)
(C) FVC%p (%)
(D) PEF%p (%)
5-year survival (95% CI)
% free of ventilation within 2 years (95% CI)
FVC (L)
PEF (L/Min)
FVC (% of predicted)
PEF (% of predicted)

Figure 3. Kaplan-Meier plot showing time from crossing 50%p to death for FVC%p (A) and PEF%p (B) and time to initiation of assisted ventilation for FVC%p (C) and PEF%p (D) in patients with decline rates <5% per year (blue line) versus ≥5% per year (red line).

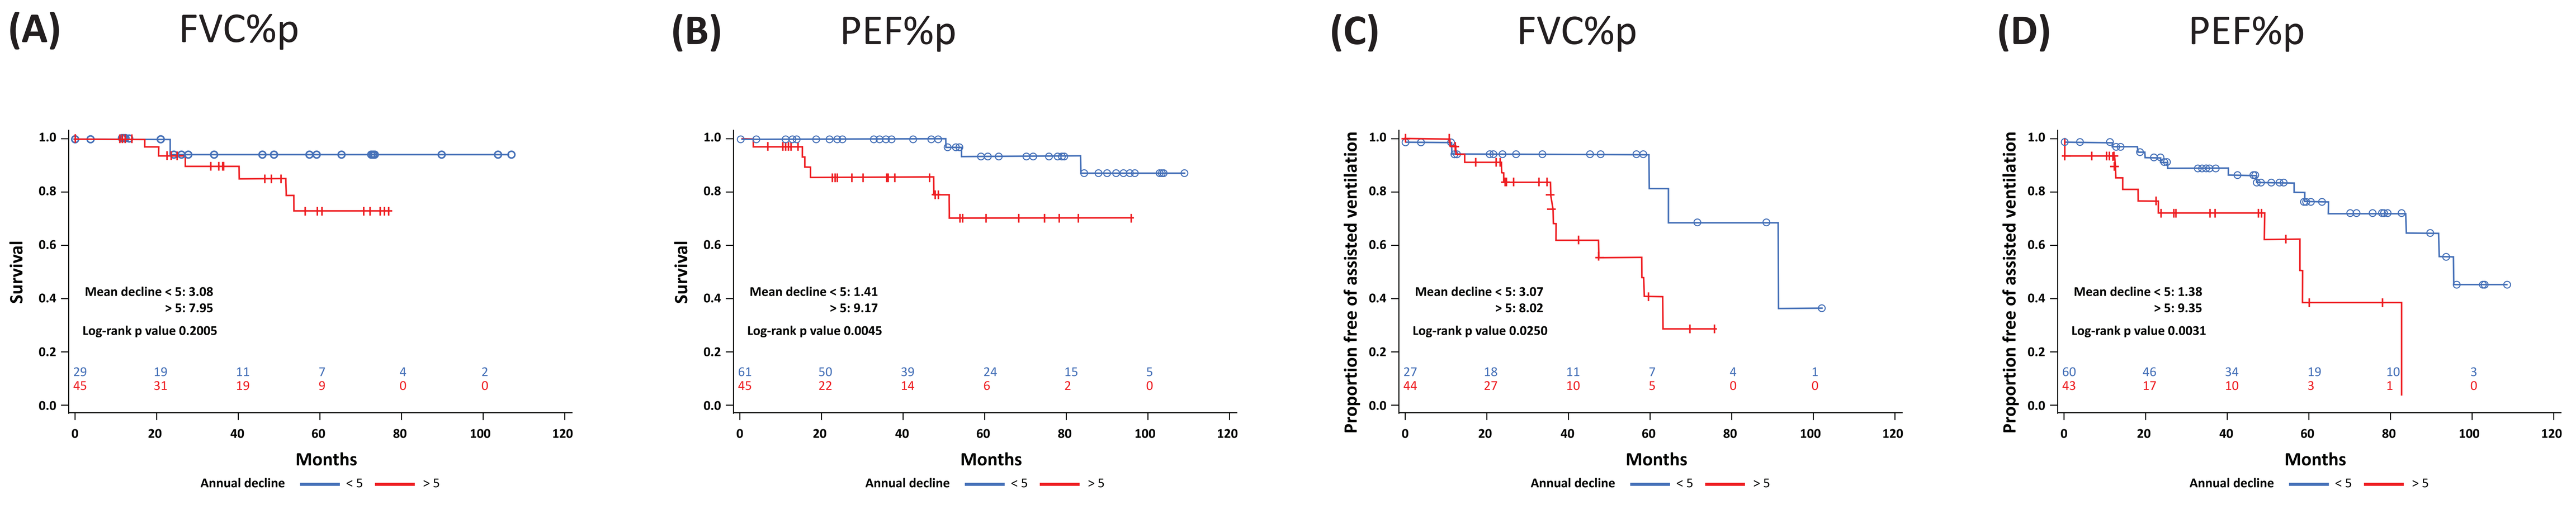
(A) FVC%p
Mean decline < 5: 3.08
> 5: 7.95
Log-rank p value 0.2005
(B) PEF%p
Mean decline < 5: 1.41
> 5: 9.17
Log-rank p value 0.0045
(C) FVC%p
Mean decline < 5: 3.07
> 5: 8.02
Log-rank p value 0.0250
(D) PEF%p
Mean decline < 5: 1.38
> 5: 9.35
Log-rank p value 0.0031
Survival
Proportion free of assisted ventilation
Months
Annual decline < 5 > 5

- 5-year estimated survival after crossing FVC of 1L was 86% (**Figure 2A**, dashed red line) and was equivalent to crossing PEF of 96L/min (**Figure 2B**, dashed red line).
- FVC%p and PEF%p predict the risk of initiation of assisted ventilation after crossing thresholds equally well (**Figure 2C and 2D**).
- There was a 3-year delay in time from crossing 50%p to initiation of assisted ventilation in <5% annual decline group (8 years) vs in >5% annual decline group (5 years) for both FVC%p and PEF%p (**Figure 3C and 3D**).
- Delay was seen also in time to death for <5% annual decline group (**Figure 3A and 3B**).
- Sensitivity analyses using cut-offs of 4%, 6% and 7% as well as crossing thresholds of 45%p, 55%p and 60%p were consistent with the primary analysis using cut-off of 5% and baseline of 50%p (data not shown).

Conclusions

- These analyses demonstrate that crossing thresholds of FVC and PEF are predictive of 5-year survival and 2-year risk of initiating assisted ventilation.
- Altering the rate of respiratory function decline impacted both the time to death as well as initiation of assisted ventilation, with a reduction in the rate of decline leading to a beneficial delay in both.
- Reducing the rate of respiratory function decline and delaying the time to cross clinically relevant thresholds of respiratory function is an important therapeutic goal to reduce respiratory complications and delaying time to needing assisted ventilation.

References

1. McDonald CM, et al. *Neuromusc Disord*, 2018:28:897-909.
2. Mayer OH, et al. *US Neurology,* 2017;13:35–41.
3. Birnkrant DJ, et al. *Lancet Neurol*, 2018;17:445-55.
4. Phillips MF, et al. *Am J Respir Crit Care Med*, 2001;164:2191-4.
5. McDonald CM, et al. *Lancet*, 2018;391:451-61.

Conflict of interest

M. Leinonen is a part-time employee of Santhera Pharmaceuticals.

Acknowledgements

CINRG DNHS participants.